Cisplatin Dose-Dependent Effect on Nausea Control: Subset Analysis from a Phase 3 Trial of NEPA versus Aprepitant/Granisetron

MASCC Annual Meeting
June 22-24, 2017

Karin Jordan[1], Corinna Lanzarotti[2], Silvia Olivari[2], Paul J Hesketh[3]

[1]Department of Medicine V, University of Heidelberg, Heidelberg, Germany; [2]Helsinn Healthcare SA, Lugano, Switzerland;
[3]Lahey Health Cancer Institute, Lahey Hospital & Medical Center, Burlington, MA

BACKGROUND

- Cisplatin is well-established as the most emetogenic chemotherapeutic agent, with antiemetic trials demonstrating a dose-dependent effect on emesis control [1-4].
- As cisplatin is unequivocally classified as highly emetogenic chemotherapy (HEC), current antiemetic guidelines do not further differentiate its emetogenicity based on this dose-dependent effect [5-7].
- In addition, antiemetic trials which frequently utilize cisplatin as the gold-standard chemotherapeutic for evaluating new antiemetic therapies, do not standardly require the same cisplatin dose threshold for patient inclusion [8-12].
- Antiemetic guidelines recommend prophylaxis with a triplet combination of an NK_1 receptor antagonist (RA), a $5\text{-}HT_3$ RA and dexamethasone (DEX) in the cisplatin-based HEC setting at all doses.
- In the first head-to-head study comparing NK1-containing regimens, a single oral dose of NEPA (fixed combination of the NK_1 RA, netupitant and $5\text{-}HT_3$ RA, palonosetron) plus DEX was similarly effective as a 3-day oral aprepitant/granisetron/DEX triplet regimen in preventing CINV in patients with various solid tumors receiving cisplatin (See MASCC oral presentation, 24 June, PS049).
- When guideline-recommended antiemetics are administered, emesis can be controlled in the majority of patients undergoing emetogenic chemotherapy. However, optimal control of nausea remains a clinical challenge [13]. Therefore, exploration of a cisplatin dose-dependent effect on nausea control was of interest, particularly as this has not been previously evaluated.

METHODS

OBJECTIVE

- This post-hoc analysis was performed from a Phase 3 study comparing NEPA + DEX versus aprepitant + granisetron + DEX to determine if nausea control differed by cisplatin dose.

STUDY DESIGN AND TREATMENT GROUPS

- This was a Phase 3, multicenter, randomized, double-blind/double-dummy, parallel group international study conducted in Asia, with the majority (81%) of patients enrolled in China.
- Patients were stratified by gender and randomly assigned (1:1) to receive either NEPA or APR/GRAN treatment **(Table 1)**.

Table 1. Treatment Groups

	NEPA Regimen		APR/GRAN Regimen		
	Oral NEPA	Oral DEX	Oral APR	IV GRAN	Oral DEX
Day 1	NETU 300 mg/ PALO 0.50 mg	12 mg	125 mg	3 mg	12 mg
Day 2		8 mg	80 mg		8 mg
Day 3		8 mg	80 mg		8 mg
Day 4		8 mg			8 mg

APR, aprepitant; **GRAN**, granisetron; **DEX**, dexamethasone; **NETU**, netupitant; **IV**, intravenous ; **PALO**, palonosetron

- NEPA and APR were administered 60 min prior to chemotherapy on day 1, while GRAN and DEX were administered 30 min prior to chemotherapy on day 1. On days 2-3, APR was administered 24 and 48h after its administration on day 1. On days 2-4, DEX was administered 24, 48 and 72h after its administration on day 1.
- The 3 mg granisetron dose is the registered dose in China.

KEY INCLUSION CRITERIA

- Males or females ≥18 years, diagnosed with a malignant solid tumor, ECOG Performance Status of 0-2, and naïve to chemotherapy
- Scheduled to receive first course of cisplatin-based (≥ 50 mg/m²) chemotherapy (as monotherapy or in combination with other chemotherapy)

KEY EXCLUSION CRITERIA

- Scheduled to receive: (i) MEC or HEC from days 2–5 following cisplatin, (ii) moderately or highly emetogenic radiotherapy within 1 week prior to day 1 or between days 1 and 5, or (iii) a bone marrow or stem-cell transplant
- Receipt of any drug with potential antiemetic efficacy within 24h before day 1
- Experienced any vomiting, retching or mild nausea within 24h before day 1
- No serious cardiovascular disease history or predisposition to cardiac conduction abnormalities with the exception of incomplete right bundle branch block
- Chronic use of select CYP3A4 inducers within 4 weeks or chronic use of a substrate or inhibitor within 1 week prior to day 1

EFFICACY/NAUSEA ASSESSMENTS

- From the start of chemotherapy on day 1 through day 5 (0-120 h), each patient completed a diary, capturing emetic episodes, severity of nausea and rescue medications intake.
- Severity of nausea was evaluated using a 100-mm horizontal visual analog scale (VAS). The left end of the scale (0 mm) was labeled as 'no nausea' and the right end of the scale (100 mm) was labeled 'nausea as bad as it could be'.
- No significant nausea (NSN) was defined as a maximum score <25 mm.
- In addition to NSN, complete response (CR: no emesis, no rescue medication) and no emesis were also assessed.

STATISTICAL ANALYSIS

- The primary focus of this post-hoc analysis was to evaluate the NSN rates for subsets of patients receiving cisplatin at doses of ≥70 mg/m² or <70 mg/m² and treated with either oral NEPA (day 1) or the 3-day oral APR/GRAN regimen.
- No formal statistical comparisons were performed to assess the efficacy between low and high cisplatin dose groups or between treatment groups.

RESULTS

BASELINE CHARACTERISTICS

- A total of 828 patients were part of this post-hoc analysis, with approximately 60% of patients receiving cisplatin ≥70 mg/m² and 40% of patients receiving cisplatin at doses <70 mg/m².
- Baseline characteristics were similar and generally balanced between high and low dose cisplatin subsets **(Table 2)**.

Table 2. Patient Baseline and Disease Characteristics (Cisplatin Dose Subsets)

	Cisplatin ≥70 mg/m²		Cisplatin <70 mg/m²	
Characteristic	**NEPA+DEX** (N = 251)	**APR/GRAN+DEX** (N = 239)	**NEPA+DEX** (N = 161)	**APR/GRAN+DEX** (N = 177)
Gender				
Male	**179** (71.3%)	**175** (73.2%)	**112** (69.6%)	**122** (68.9%)
Female	**72** (28.7%)	**64** (26.8%)	**49** (30.4%)	**55** (31.1%)
Age (years), Mean	**53.9**	**53.8**	**55.6**	**55.5**
ECOG Performance Status				
0	**100** (39.8%)	**88** (36.8%)	**75** (46.6%)	**83** (46.9%)
1	**148** (59.0%)	**149** (62.3%)	**82** (50.9%)	**87** (49.2%)
2	**3** (1.2%)	**2** (0.8%)	**4** (2.5%)	**7** (4.0%)
Most Common (≥ 5%) Cancer Types				
Lung	**167** (66.5%)	**152** (63.6%)	**108** (67.0%)	**115** (65.0%)
Head and neck	**50** (19.9%)	**52** (21.8%)	**10** (6.2%)	**14** (7.9%)
Cisplatin Dose (mg/m²)				
Mean	**77.2**	**77.5**	**61.8**	**61.2**
Median	**75**	**75**	**62.7**	**61.3**
Most Common (≥ 5%) Concomitant Chemotherapy				
Gemcitabine	**82** (32.7%)	**58** (24.3%)	**41** (25.5%)	**35** (19.8%)
Pemetrexed	**47** (18.7%)	**54** (22.6%)	**22** (13.7%)	**25** (14.1%)
Docetaxel	**32** (12.7%)	**31** (13.0%)	**35** (21.7%)	**37** (20.9%)
Etoposide	**33** (13.1%)	**25** (10.5%)	**25** (15.5%)	**28** (15.8%)
Fluorouracil	**20** (8.0%)	**26** (10.9%)	**7** (4.3%)	**6** (3.4%)

APR, aprepitant; **GRAN**, granisetron; **DEX**, dexamethasone; **SD**, standard deviation; **ECOG**, Eastern Cooperative Oncology Group

EFFICACY

- A cisplatin dose-dependent effect on nausea control was shown in both groups, although to a greater extent for APR/GRAN, where ~10% fewer patients had NSN in the high dose cisplatin group compared with the low dose group during the delayed and overall phases **(Figure 1)**.
- Numerically higher NSN rates were seen for NEPA than APR/GRAN during all phases but, in particular, in the high dose cisplatin group.

Figure 1. NSN Rates: Cisplatin Dose Subsets

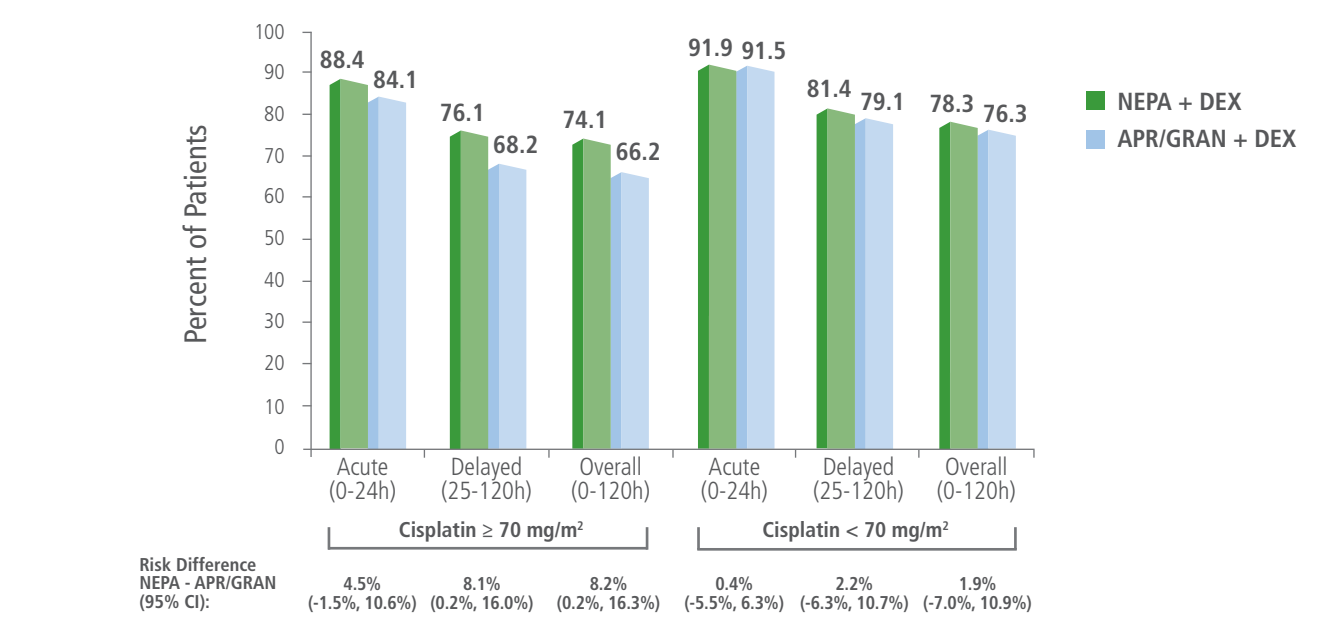
Percent of Patients
100
90
80
70
60
50
40
30
20
10
0
88.4
84.1
76.1
68.2
74.1
66.2
91.9
91.5
81.4
79.1
78.3
76.3
Acute (0-24h)
Delayed (25-120h)
Overall (0-120h)
Acute (0-24h)
Delayed (25-120h)
Overall (0-120h)
Cisplatin ≥ 70 mg/m²
Cisplatin < 70 mg/m²
NEPA + DEX
APR/GRAN + DEX
Risk Difference NEPA - APR/GRAN (95% CI):
4.5% (-1.5%, 10.6%)
8.1% (0.2%, 16.0%)
8.2% (0.2%, 16.3%)
0.4% (-5.5%, 6.3%)
2.2% (-6.3%, 10.7%)
1.9% (-7.0%, 10.9%)

- A similar dose-dependent effect of cisplatin was seen for no emesis and complete response rates **(Table 3)**.

Table 3. No Emesis and Complete Response Rates: Cisplatin Dose Subsets

	Cisplatin ≥70 mg/m²		Cisplatin <70 mg/m²	
% of Patients	**NEPA** N=251	**APR/GRAN** N=239	**NEPA** N=161	**APR/GRAN** N=177
No Emesis				
Acute	**82.1%**	**84.5%**	**90.1%**	**91.5%**
Delayed	**77.7%**	**71.1%**	**82.0%**	**83.1%**
Overall	**72.5%**	**68.2%**	**78.9%**	**81.9%**
Complete Response				
Acute	**81.3%**	**83.7%**	**89.4%**	**91.5%**
Delayed	**75.7%**	**69.0%**	**81.4%**	**81.4%**
Overall	**70.9%**	**66.5%**	**78.3%**	**80.2%**

CONCLUSIONS

- Prevention of nausea, particularly during the delayed phase following HEC, should remain a priority of current antiemetic research.
- Triplet antiemetic prophylaxis prevented significant nausea in the majority of patients in this study, and was numerically more frequent with NEPA during the delayed and overall phases in the subset of patients receiving high dose cisplatin.
- Consistent with prior reports for emesis, in this study comparing guideline-recommended antiemetic regimens, a higher cisplatin dose was associated with lower rates of nausea control, confirming the importance of cisplatin dose in predicting nausea development.

ACKNOWLEDGEMENTS: The authors would like to thank the clinical investigators, patients and site personnel at each of the participating centers. Editorial and medical writing assistance was provided by Jennifer Vanden Burgt, Minneapolis, MN and funded by Helsinn Healthcare SA, Lugano, Switzerland. The trial was sponsored by Helsinn Healthcare SA.

REFERENCES: **1.** Hesketh PJ, et al. Supp Care Cancer 2010; 18: 1171-1177. **2.** duBois A, et al. Eur J Cancer 1992; 28(213): 450-457. **3.** Hellenbrecht D, Saller R. Arzneimittelforschung 1986; 36(12):1845-9. **4.** Pritchard JF, Wells CD. Pharmacology 1992; 45(4):188-94. **5.** Hesketh PJ, et al. J Clin Oncol 2016;34(4):381-6. **6.** Roila F, et al. Ann Oncol 2016;27(5):v119-v133. **7.** National Comprehensive Cancer Network. NCCN clinical practice guidelines in oncology: Antiemesis [v1.2017]. https://www.nccn.org Accessed March 2017. **8.** Hesketh PJ, et al. Ann Oncol 2014;25:1340-6. **9.** Poli-Bigelli S, et al. Cancer 2003;97(12):3090-3098. **10.** Hesketh PJ, et al. J Clin Oncol 2003;21(22):4112-4119. **11.** Schmoll HJ, et al. Ann Oncol 2006;17(6):1000-1006. **12.** Rapoport B, et al. Lancet Oncol 2015;16(9):1079-1089. **13.** Bosnjak S, et al. Support Care Cancer 2017;25(5):1661-1671.