Pooled Analysis of Two Phase 3 Trials: Body Weight Response with Anamorelin in Advanced Non-small Cell Lung Cancer (NSCLC) Patients with Anorexia/Cachexia

MASCC 2017
Abstract eP348

David Currow[1], Jennifer Temel[2], Amy Abernethy[3], John Friend[4], Ruben Giorgino[5]

[1]ImPACCT – Improving Palliative Aged and Chronic Care through Clinical Research and Translation, Faculty of Health, University of Technology Sydney, Sydney (NSW), Australia; [2]Massachusetts General Hospital, Boston (MA), USA; [3]Flatiron Health, New York (NY), USA; [4]Cellectar Biosciences, Madison (WI), USA; [5]Helsinn Healthcare SA, Lugano, Switzerland

INTRODUCTION

Cancer Anorexia/Cachexia

- A frequent, debilitating condition, characterized by ongoing weight loss (mostly due to loss of lean body mass [LBM]), driven by a combination of reduced food intake and abnormal metabolism.[1]
 - The most recent international consensus[1] suggests the following diagnostic criteria:
 - Weight loss ≥5%, or
 - Weight loss ≥2% and body mass index (BMI) <20 kg/m^2, or
 - Weight loss ≥2% in patients with sarcopenia
- Leads to progressive functional impairment, and is associated with reduced quality of life, decreased tolerance/response to chemotherapy, and augmented morbidity/mortality.[2,3]
- Occurs in approximately 70% of patients with advanced cancers,[4] and has a high prevalence in patients with NSCLC.[5]
- Currently available therapeutic options for cancer anorexia/cachexia have limited efficacy and are associated with possible safety risks, specifically in patients with advanced cancers.[6]

Anamorelin HCl

- Ghrelin, the endogenous ligand of the ghrelin receptor, stimulates multiple pathways involved in regulation of appetite, body weight, LBM, and metabolism.[7]
- Anamorelin is a novel, highly selective, orally active ghrelin receptor agonist that presents similar appetite-enhancing and anabolic properties to those of ghrelin, thereby enabling energy storage.[8]
- Efficacy and safety of anamorelin over 12 weeks have been evaluated in the international, randomized, double-blind phase 3 trials ROMANA 1 (NCT01387269) and ROMANA 2 (NCT01387282) in patients with advanced NSCLC and cachexia.[9]
 - Anamorelin was well tolerated and significantly increased LBM and other body composition parameters, compared with placebo
 - Anamorelin, versus placebo, also significantly improved anorexia/cachexia symptom burden, while no differences in handgrip strength (HGS) were observed

OBJECTIVE

- Considering that involuntary weight loss of ≥5% is an established diagnostic criterion for anorexia/cachexia, this analysis assessed the proportions of patients with ≥5% increase in body weight at the end of study (EOS), following anamorelin treatment.
 - This analysis was performed in the overall modified intent-to-treat (mITT) population, and in patients with BMI <20 kg/m^2 at baseline (who met the cachexia definition within the inclusion criteria)

MATERIALS AND METHODS

Study Design

- ROMANA 1 and ROMANA 2 were two international, double-blind, randomized, placebo-controlled phase 3 trials, for which full eligibility criteria and primary results have been previously reported.[9]
- Patients with unresectable stage III/IV NSCLC and cachexia (≥5% body weight loss during the prior 6 months, or BMI <20 kg/m^2 at baseline) were randomized (2:1) to receive 100 mg once-daily oral anamorelin or placebo tablets for up to 12 weeks.
- Patients could receive concomitant chemotherapy, but not concomitant medications for treating weight loss or for increasing appetite.
- This pooled post-hoc analysis assessed:
 - The efficacy of anamorelin and the proportions of patients with an increase in body weight ≥5% at EOS (or last observation carried forward since week 6 or 9), in the overall mITT population and in patients with BMI <20 kg/m^2 at baseline

Statistical Analyses

- A post-hoc analysis of pooled efficacy data from ROMANA 1 and ROMANA 2 was conducted.
- The efficacy analyses were performed on the overall mITT population (defined as all randomized patients who received any study drug and have had more than 1 post-baseline LBM or HGS measurement).
- Data were described by mean and descriptive statistics.
 - Changes from baseline to EOS, 95% confidence intervals (CIs), percentages of patients with an increase in body weight ≥5% at EOS, and nominal p values were reported

RESULTS

Patient Population

- The pooled analysis contained a total of 829 patients (**Figure 1**) in the overall mITT population.

Figure 1. Pooled ROMANA 1 and ROMANA 2 Efficacy Analysis: Patient Disposition

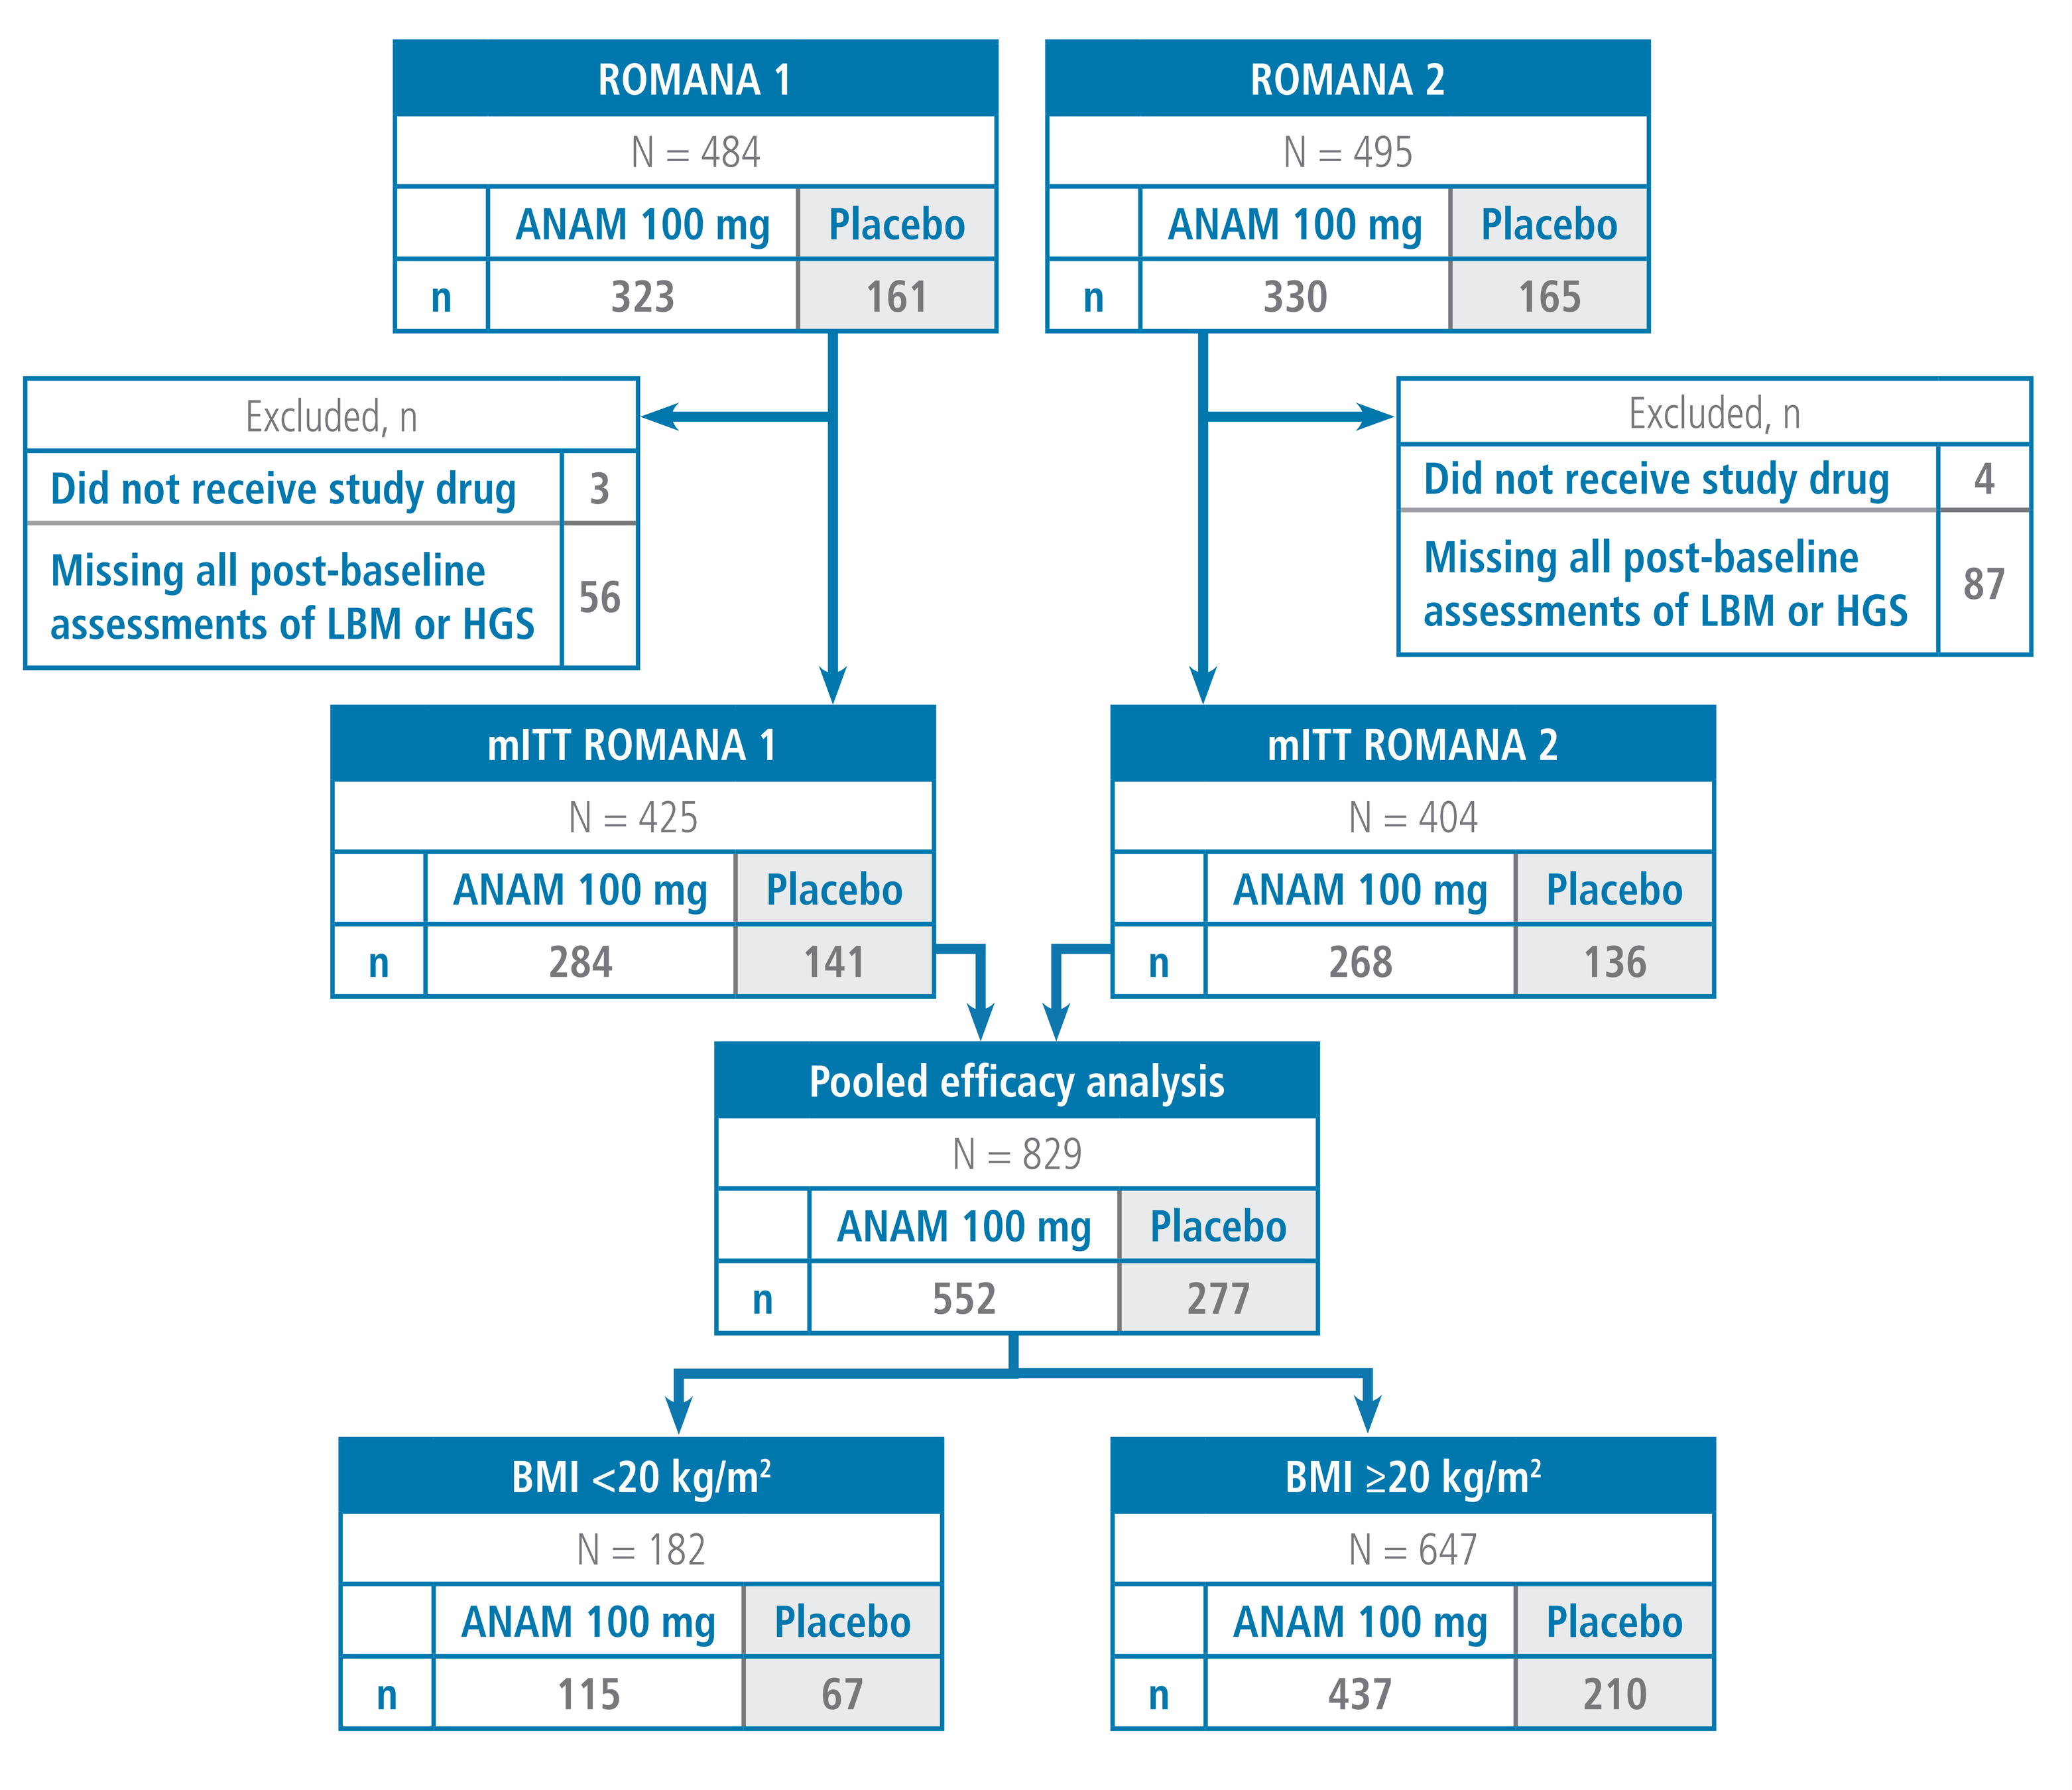
ROMANA 1
N = 484
ANAM 100 mg
Placebo
n
323
161
ROMANA 2
N = 495
ANAM 100 mg
Placebo
n
330
165
Excluded, n
Did not receive study drug
3
Missing all post-baseline assessments of LBM or HGS
56
Excluded, n
Did not receive study drug
4
Missing all post-baseline assessments of LBM or HGS
87
mITT ROMANA 1
N = 425
ANAM 100 mg
Placebo
n
284
141
mITT ROMANA 2
N = 404
ANAM 100 mg
Placebo
n
268
136
Pooled efficacy analysis
N = 829
ANAM 100 mg
Placebo
n
552
277
BMI <20 kg/m2
N = 182
ANAM 100 mg
Placebo
n
115
67
BMI ≥20 kg/m2
N = 647
ANAM 100 mg
Placebo
n
437
210

ANAM: anamorelin HCl; **BMI:** body mass index; **HGS:** handgrip strength; **LBM:** lean body mass; **mITT:** modified intent-to-treat.

- Baseline demographics and clinical characteristics of patients are summarized in **Table 1**.

Table 1. Patient Demographics and Baseline Characteristics

	Overall mITT population (N = 829)		Baseline BMI <20 kg/m^2 (N = 182)	
	ANAM 100 mg (N = 552)	Placebo (N = 277)	ANAM 100 mg (N = 115)	Placebo (N = 67)
Female, n (%)	134 (24.3)	75 (27.1)	31 (27.0)	22 (32.8)
Age, years, mean (SD)	62.2 (9.0)	62.4 (8.8)	60.3 (9.1)	62.5 (9.1)
Metastatic disease at study entry, n/N (%)	408/551 (74.0)	189/276 (68.5)	78/115 (67.8)	50/66 (75.8)
Time from initial tumor diagnosis, months, median (IQR)	8.4 (19.8)	8.0 (14.5)	7.2 (12.7)	9.7 (13.1)
Chemo/radiotherapy to be started within 14 days, n (%)	474 (85.9)	235 (84.8)	99 (86.1)	47 (70.1)
Concomitant use of opioids, n (%)	166 (30.1)	81 (29.2)	60 (52.2)	30 (44.8)
Body weight, kg, mean (SD)	66.78 (13.06)	65.76 (13.50)	52.08 (6.84)	51.62 (8.04)
LBM, kg, mean (SD)	45.44 (8.02)	45.05 (8.73)	40.17 (6.65)	39.54 (6.79)
aLBM, kg, mean (SD)	19.32 (4.20)	19.12 (4.40)	16.36 (3.37)	16.15 (3.39)
FM, kg, mean (SD)	18.87 (8.13)	18.59 (8.02)	9.75 (3.27)	10.26 (3.46)
FAACT A/CS score, mean (SD)	29.45 (8.44)	30.02 (8.38)	25.33 (8.59)	27.77 (7.96)
FACIT-F score, mean (SD)	30.35 (10.42)	30.69 (10.61)	27.51 (11.69)	30.31 (10.99)

aLBM: appendicular lean body mass; **ANAM:** anamorelin HCl; **BMI:** body mass index; **FAACT A/CS:** Functional Assessment of Anorexia/Cachexia Therapy – Anorexia/Cachexia Subscale; **FACIT-F:** Functional Assessment of Chronic Illness Therapy – Fatigue; **FM:** fat mass; **IQR:** interquartile range; **LBM:** lean body mass; **mITT:** modified intent-to-treat; **SD:** standard deviation.

Efficacy

- In the mITT population, treatment with anamorelin led to a significant increase in body weight (**Figure 2A**) and in the percentage of patients with an increase in body weight ≥5% at EOS (**Figure 2B**).
- In patients with BMI <20 kg/m^2 at baseline, anamorelin led to greater improvements in body weight when compared with placebo (**Figure 3A**).
 - A significantly higher percentage of patients with an increase in body weight ≥5% at EOS was observed following anamorelin treatment, versus placebo (**Figure 3B**)
- The proportion of patients benefiting from anamorelin treatment was higher in patients with BMI <20 kg/m^2 at baseline (47.3%) than in the mITT population (34.1%).

Figure 2. Body Weight in the Overall mITT Population: A) Change from Baseline to EOS in Body Weight, and B) Proportions of Patients with an Increase in Body Weight ≥5% at EOS, per Treatment Arm

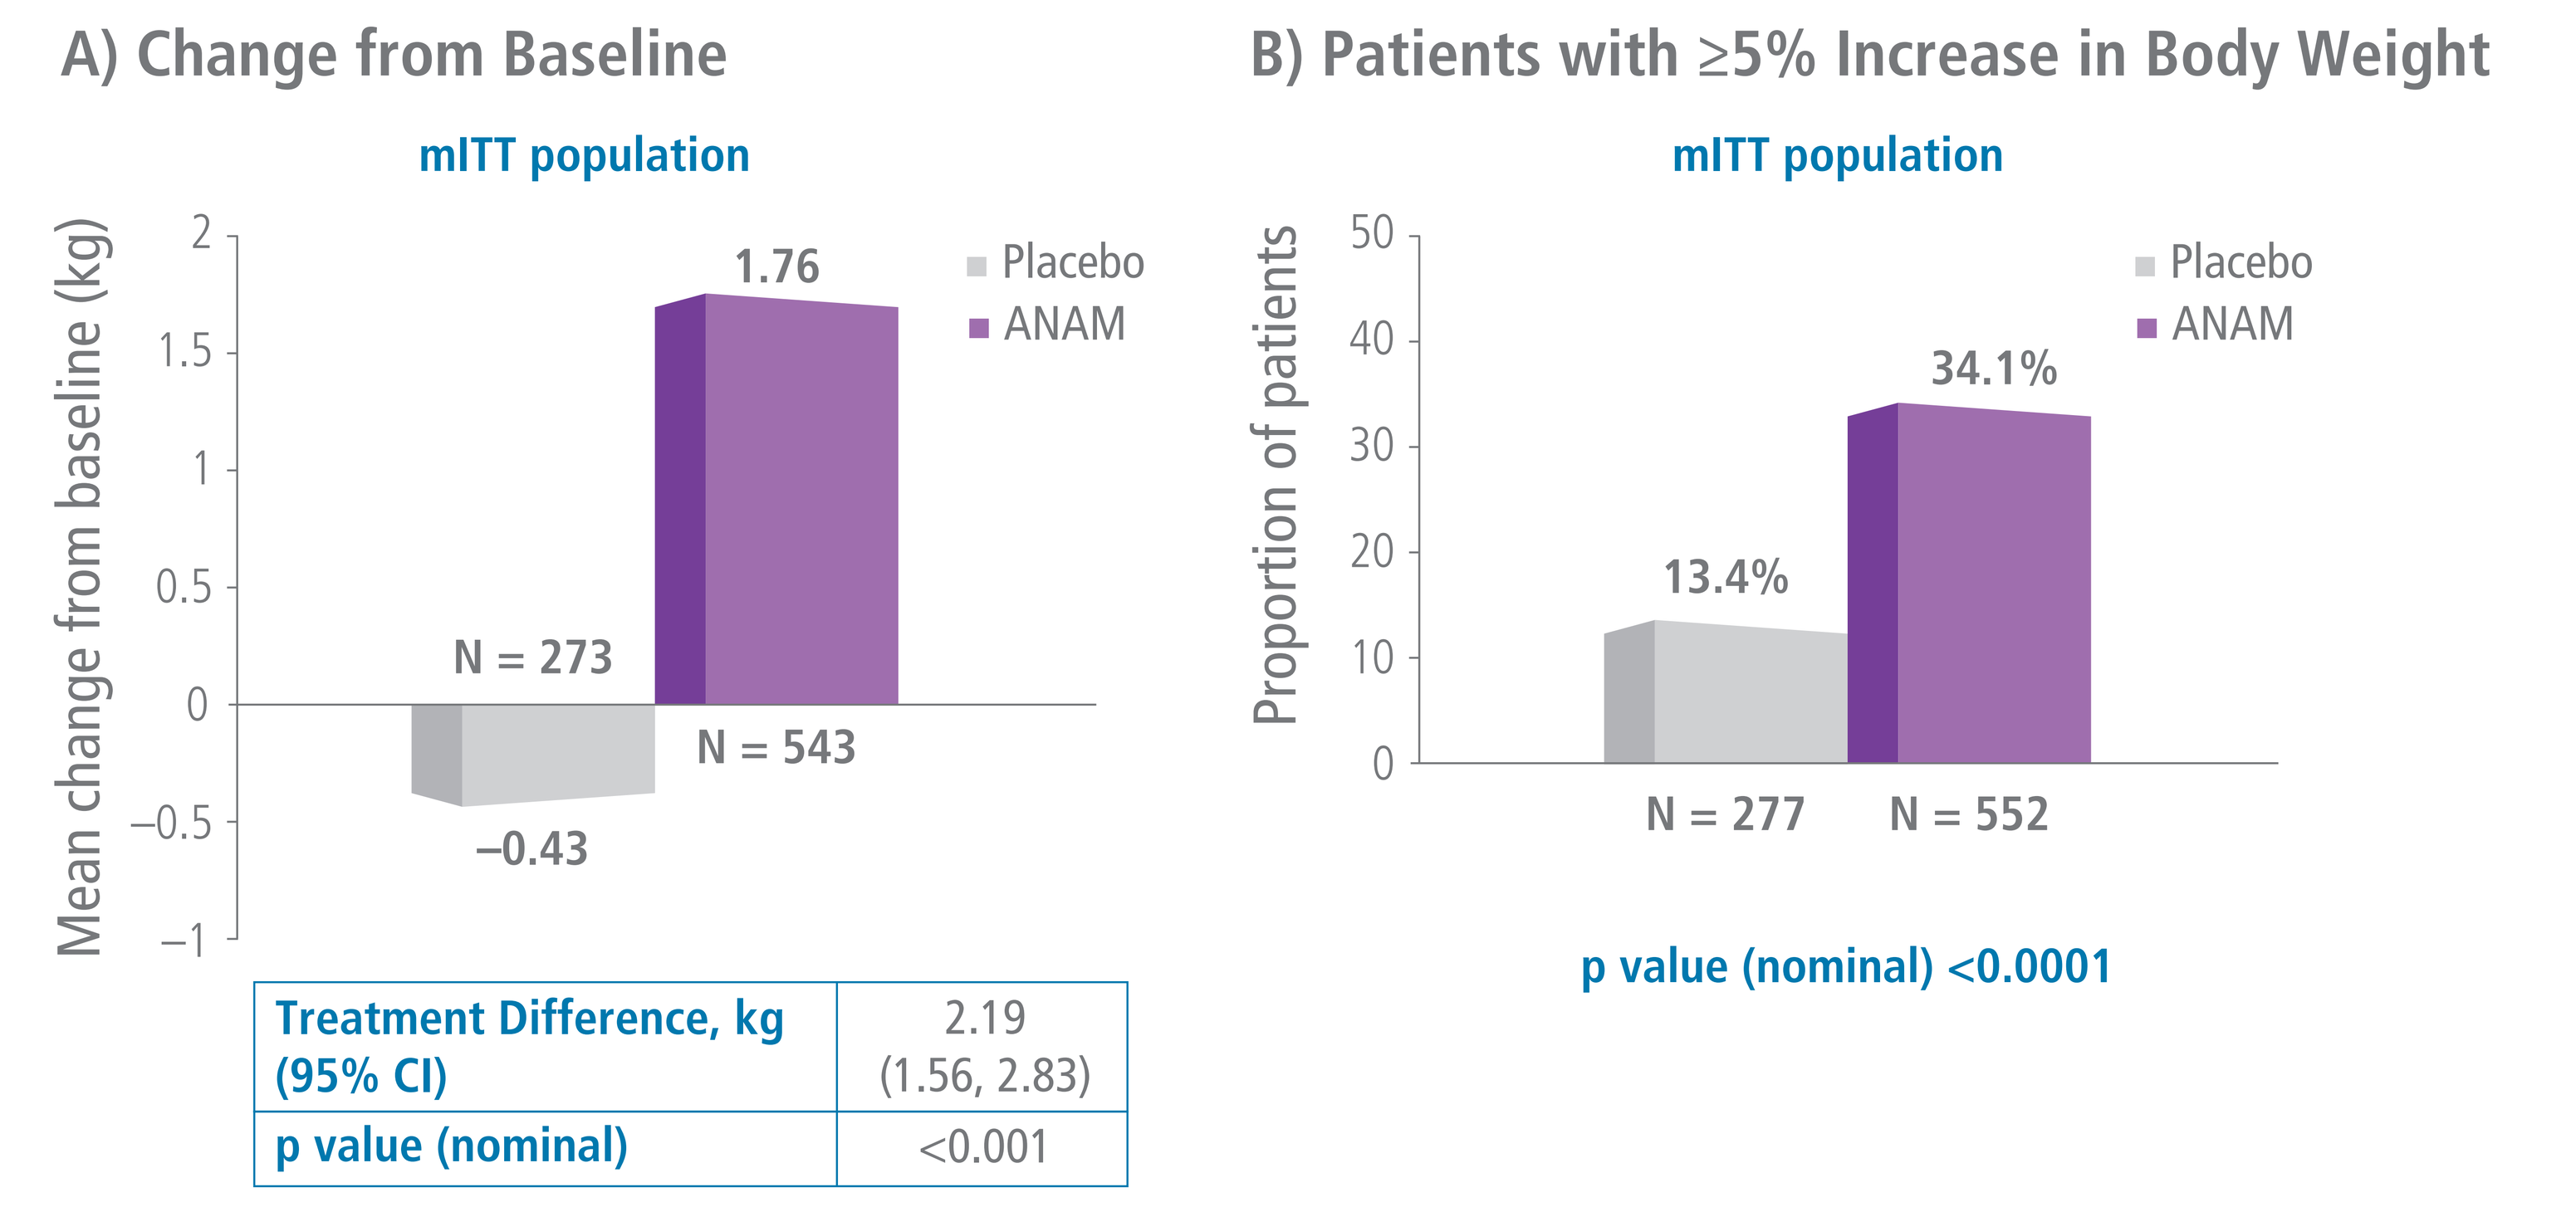
A) Change from Baseline
mITT population
Mean change from baseline (kg)
2
1.5
1
0.5
0
–0.5
–1
Placebo
ANAM
1.76
N = 273
N = 543
–0.43
B) Patients with ≥5% Increase in Body Weight
mITT population
Proportion of patients
50
40
30
20
10
0
Placebo
ANAM
34.1%
13.4%
N = 277
N = 552
p value (nominal) <0.0001

Treatment Difference, kg (95% CI)	2.19 (1.56, 2.83)
p value (nominal)	<0.001

Mean change from baseline to EOS in body weight is presented in patients that have had all three study measurements (week 6, week 9, and EOS). The proportion of patients with ≥5% increase in body weight at EOS is presented in the overall mITT population.
ANAM: anamorelin; **CI:** confidence interval; **EOS:** end of study; **mITT:** modified Intent-to-treat.

Figure 3. Body Weight in Patients with BMI <20kg/m^2 at Baseline: A) Change from Baseline to EOS in Body Weight, and B) Proportions of Patients with an Increase in Body Weight ≥5% at EOS, per Treatment Arm

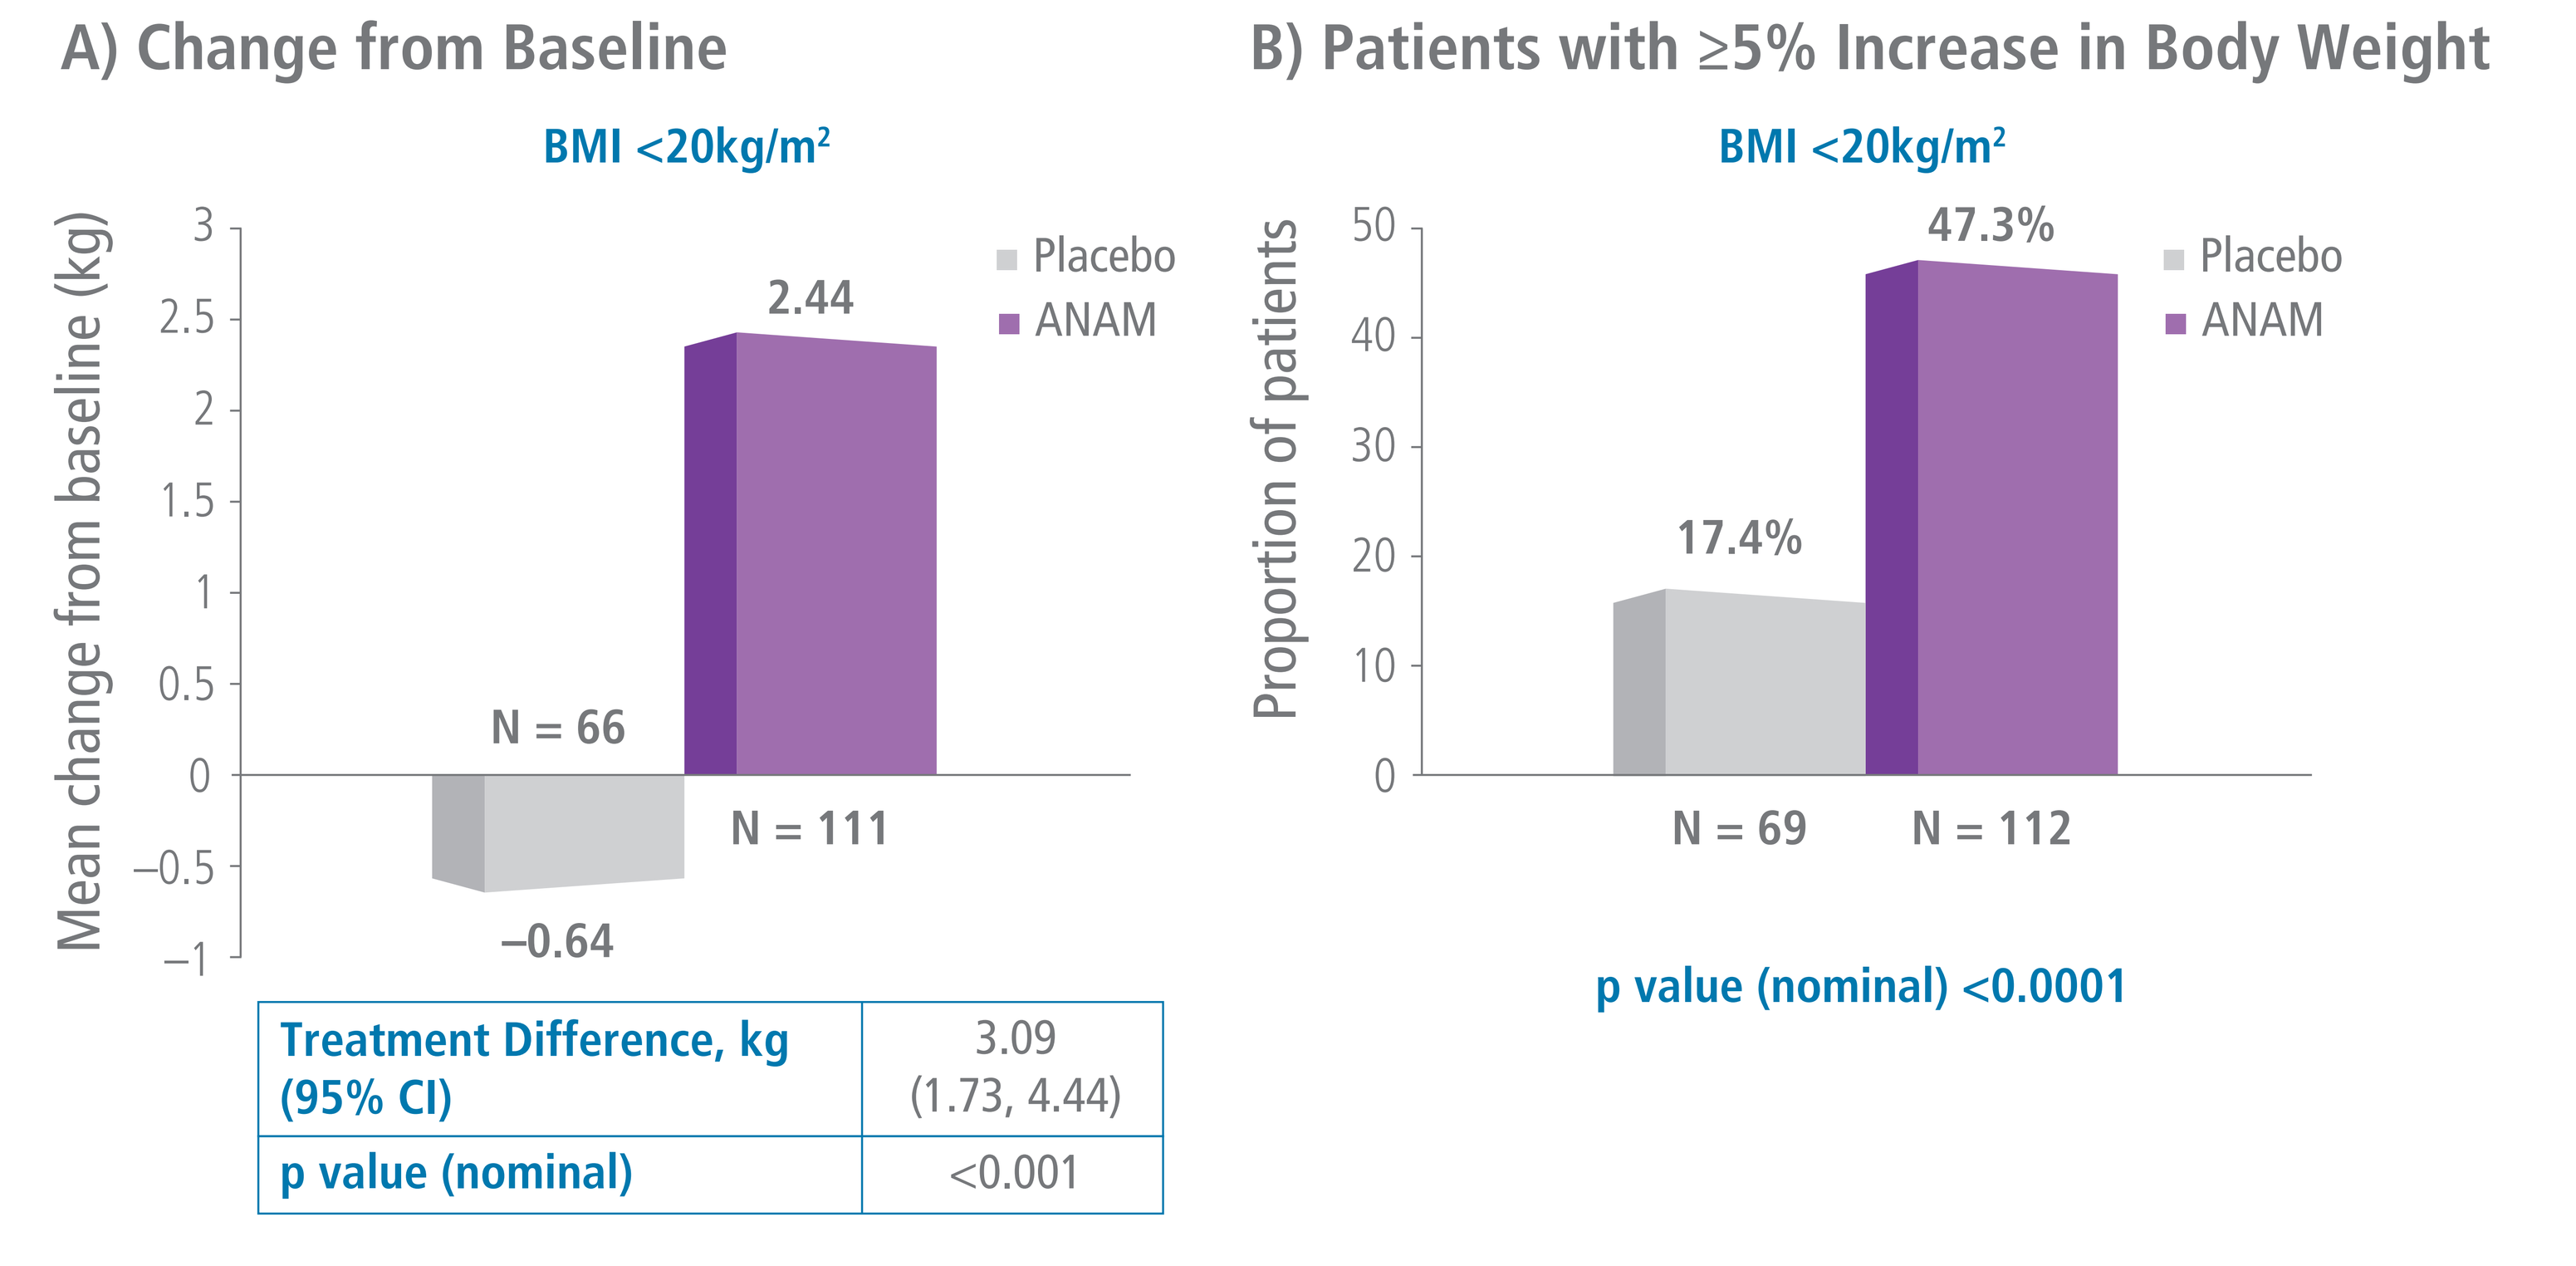
A) Change from Baseline
BMI <20kg/m2
Mean change from baseline (kg)
3
2.5
2
1.5
1
0.5
0
–0.5
–1
Placebo
ANAM
2.44
N = 66
N = 111
–0.64
B) Patients with ≥5% Increase in Body Weight
BMI <20kg/m2
Proportion of patients
50
40
30
20
10
0
Placebo
ANAM
47.3%
17.4%
N = 69
N = 112
p value (nominal) <0.0001

Treatment Difference, kg (95% CI)	3.09 (1.73, 4.44)
p value (nominal)	<0.001

Mean change from baseline to EOS in body weight is presented in patients that have had all three study measurements (week 6, week 9, and EOS). The proportion of patients with ≥5% increase in body weight at EOS is presented in all patients with BMI <20kg/m^2 at baseline.
ANAM: anamorelin; **BMI:** body mass index; **CI:** confidence interval; **EOS:** end of study.

CONCLUSIONS

- The results of the ROMANA 1 and ROMANA 2 phase 3 trials in advanced NSCLC patients with cachexia indicate the clinical relevancy of anamorelin's treatment effect size on body composition.
 - This is shown by the higher response rate attained upon applying the stringent cutoff of ≥5% weight gain
- The high proportion of patients with BMI <20 kg/m^2 at baseline who had a body weight increase ≥5% indicates that patients with more advanced cachexia can still benefit from treatment with anamorelin.

REFERENCES: 1. Fearon K, et al. *Lancet Oncol.* 2011;12:489–95. **2.** LeBlanc TW, et al. *J Pain Symptom Manage.* 2015;49:680–9. **3.** Fearon K, et al. *Nat Rev Clin Oncol.* 2013;10:90–9. **4.** Yavuzsen T, et al. *J Clin Oncol.* 2005;23:8500–11. **5.** Del Ferraro C, et al. *J Hosp Palliat Nurs.* 2012;14. **6.** Macciò A, et al. *Expert Opin Pharmacother.* 2012;13:2453–72. **7.** Guillory B, et al. *Vitam Horm.* 2013;92:61–106. **8.** Currow DC, et al. *Future Oncol.* 2014;10:789–802. **9.** Temel JS, et al. *Lancet Oncol.* 2016;17:519–31.

ACKNOWLEDGMENTS: Kenneth C. Fearon significantly contributed to trial design and data analysis. The trials were sponsored by Helsinn Therapeutics (U.S.) Inc., Iselin (NJ), USA. The authors would like to thank patients, investigators, and study teams at the participating institutions in the following countries: Australia, Belarus, Belgium, Canada, Czech Republic, France, Germany, Hungary, Israel, Italy, The Netherlands, Poland, Russia, Serbia, Slovenia, Spain, Ukraine, UK, and USA. The authors would also like to acknowledge both Maurizio Rainisio and Enrico Baroni for their contributions. Editorial and medical writing assistance was provided by Oana Draghiciu, PhD, TRM Oncology, The Hague, The Netherlands, funded by Helsinn Healthcare SA, Lugano, Switzerland. The authors are fully responsible for all content and editorial decisions for this poster.

DISCLOSURES: DC: Consulting or advisory role, Helsinn, Mayne Pharma, and Specialised Therapeutics Australia Pty. Ltd. **JT:** None to disclose. **AA:** Consulting or advisory role, Bristol-Myers Squibb and Helsinn; stock ownership, athenahealth, Flatiron Health, Orange Leaf Associates; board of directors: athenahealth; employee of Flatiron Health; leader at athenahealth. **JF:** Employee, Cellectar Biosciences (USA). **RG:** Employee, Helsinn Healthcare SA; patents, Helsinn Healthcare SA.