Melatonin oral gel for the prevention of oral mucositis H&N cancer undergoing chemo/bioradiation (MUCOMEL)

A.Lozano[1]; J. Marruecos[2]; N.Farré[3]; R. Morera[4];I. Planas[5]; J. Giralt[6]; M. Lanzuela[7]; A. Escribano[4]; L.A. Glaria[4]; R. Mesía[1]; J. Rubió[2]; A. López-Pousa[3]; N.Basté[6]; B.Castelo[4]; B. Cirauqui[5]; J. Martínez-Trufero[7]; J. Ortíz[9]; P.M. Grima[9]; V.Valentí[8]; C.Tarragó[9]; R.Bosser[9]

[1] Inst. Català d'Oncologia L'Hospitalet, Barcelona (SPA); [2]Inst. Català d'Oncologia Girona (SPA); [3] Hosp. Universitari de la Santa Creu i Sant Pau, Barcelona (SPA); [4]Hosp. Universitario La Paz, Madrid (SPA); [5]Inst. Català d'Oncologia Badalona, Barcelona (SPA); [6]Hosp. Universitari de la Vall d'Hebron, Barcelona (SPA); [7]Hosp. Universitario Miguel Servet, Zaragoza (SPA); [8]Hosp. de Santa Tecla, Tarragona (SPA); [9]Spherium Biomed S.L., Esplugues de Llobregat, Barcelona (SPA)

ABSTRACT

The objective of this ongoing Phase Ib-II trial is to evaluate the safety of melatonin (MLT) oral gel, its efficacy in the prevention of severe Oral Mucositis (OM) in H&N cancer patients (84) and to assess the pharmacokinetic profile of MLT in the subgroup of the first 24 patients.

Materials and Methods:
This is a multicenter, prospective, randomized, double blind and placebo-controlled study. Eligible patients are assigned at 1:1 ratio to receive 3% melatonin or matching placebo oral gels (mouthwashes &swallowing).
Selected radiotherapy is VMAT-SIB once daily (5d/w), 50.4 Gy (low risk area), 69.96 Gy (high risk area) and 66 Gy (post-operative oral cavity tumour). Radiotherapy plan was previously agreed between all the radiotherapists participating in the study in order to ensure homogeneity between centres. Concurrent systemic treatment is either cisplatin or cetuximab.
The primary endpoint is to identify the num. of patients with severe OM (G3-G4/RTOG), other efficacy, QoL and safety endpoints will be analysed, as well.

Results: The first patient was enrolled in November 2015 and, up to 14 February 2017, 56 patients were randomized to treatment with either MLT or placebo oral gel. Results are expected by end of 2017.

Conclusion: This prospective, randomized, double-blind and placebo-controlled study will demonstrate whether melatonin oral gel 3% is safe and has been able to prevent severe OM in H&N cancer patients undergoing QRT, and if has shown efficacy in the other evaluated endpoints.

INTRODUCTION

Oral mucositis (OM) is the most significant adverse event (AE) in patients undergoing concurrent chemo/biotherapy plus radiotherapy for treating head and neck (H&N) cancer. OM is associated with nutritional issues, impairment of the quality of life, high economic cost and decreased efficacy of the antineoplastic treatment.
Oral mouthwashes with a melatonin 3% mucoadhesive oral gel have been shown to prevent OM in preclinical models of rats undergoing experimental radiation, and furthermore, previous early clinical experience confirmed these preclinical observations.
The objective of the ongoing Phase Ib-II trial is to evaluate the safety of melatonin (MLT) oral gel, its efficacy in the prevention of severe OM in H&N cancer patients (84) and to assess the pharmacokinetic profile of MLT in the subgroup of the first 24 patients.

METHODS

This is a multicentric, prospective, randomized, double blind and placebo-controlled study, currently ongoing in seven centers in Spain. Estimated enrollment: 84 patients.
Some of the main inclusion criteria are:

- Male and female patients ≥18 years, life expectancy ≥ 3 months, ECOG performance status 0-1
- Histologically confirmed diagnosis of non-metastatic TNM-2010 stage III-IV squamous cell carcinoma of the following sites: Oral cavity, oropharynx or any H&N site with lymph nodes at cervical level II; or histologically confirmed carcinoma of the nasopharynx (differentiated squamous cell carcinoma or NonKeratinizing carcinoma or undifferentiated carcinoma) found eligible for chemoradiation with or without neoadjuvant chemotherapy.
- Patients who have a treatment plan based on systemic treatment (cisplatin or cetuximab) concurrent with radiation with curative intent. Patients may have received up to 3 cycles of neoadjuvant chemotherapy if local adverse events related to this treatment are fully resolved before study entry. Patients with a plan of postoperative chemoradiation may be included only if the primary tumour is located in the oral cavity.

RADIOTHERAPY

Technical characteristics of the radiotherapy (RT) are homogeneous among participating centers regarding dose fractionation, volumes, source and doses of radiation and use of high precision systems such as VMAT. Radiation Oncologists approved by consensus the planning Organs at Risk Volume (PRV).

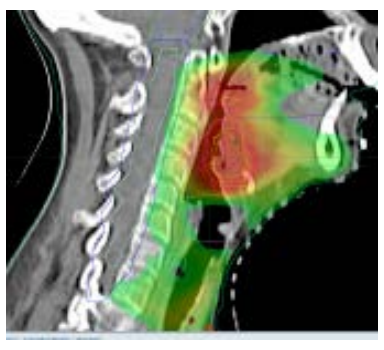

Total RT dose ≥ 66 Gy (33 sessions), with the following schedule:

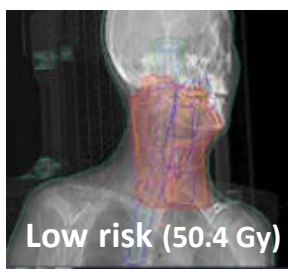
Low risk (50.4 Gy)

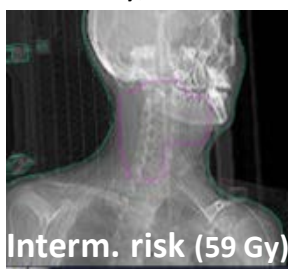
Interm. risk (59 Gy)

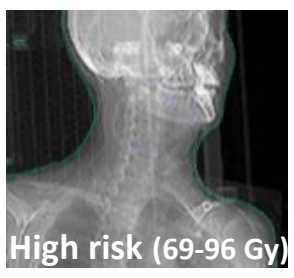
High risk (69-96 Gy)

Two mucosal volumes are designed in all patients in order to relate radiation with mucositis severity.

- Oral cavity mucosa (morphological)
- Pharyngeal mucosa (functional)

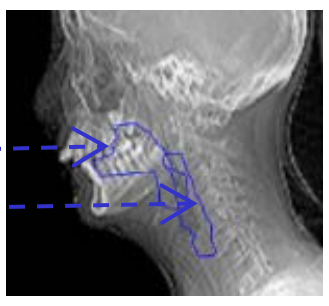

TRIAL DESIGN

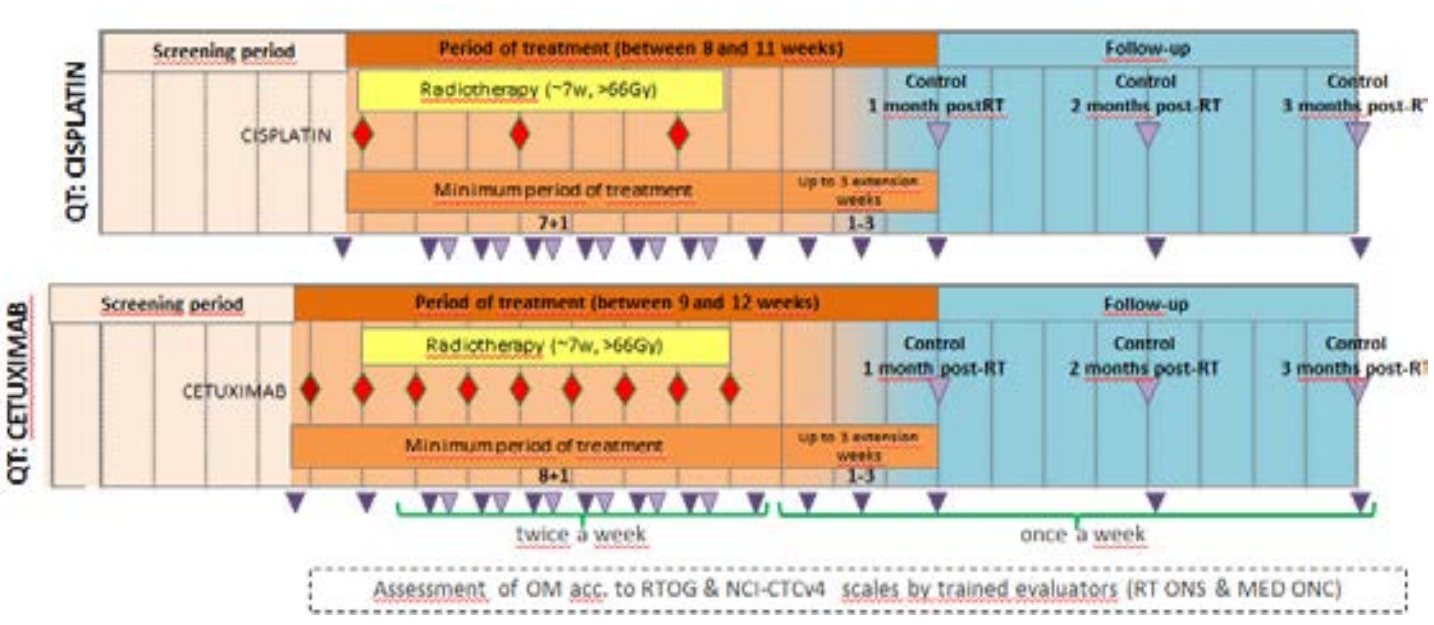
QT: CISPLATIN
Screening period
Period of treatment (between 8 and 11 weeks)
Follow-up
Radiotherapy (~7w, >66Gy)
CISPLATIN
Control 1 month postRT
Control 2 months post-RT
Control 3 months post-RT
Minimum period of treatment
7+1
Up to 3 extension weeks
1-3
QT: CETUXIMAB
Screening period
Period of treatment (between 9 and 12 weeks)
Follow-up
Radiotherapy (~7w, >66Gy)
CETUXIMAB
Control 1 month post-RT
Control 2 months post-RT
Control 3 months post-RT
Minimum period of treatment
8+1
Up to 3 extension weeks
1-3
twice a week
once a week
Assessment of OM acc. to RTOG & NCI-CTCv4 scales by trained evaluators (RT ONS & MED ONC)

CONCURRENT SYSTEMIC ANTINEOPLASTIC TREATMENT

- Cisplatin (CDDP) 100mg/m^2 on days 0, 21 & 42 of RT OR
- Cetuximab 400mg/m^2 loading dose (d -7 of RT), & 250mg/m^2 weekly during RT.

SYMPTOMATIC TREATMENT FOR OM & OTHER ADVERSE EVENTS (Aes)

All patients receive standard symptomatic treatment for OM along the study as per hospital routine clinical practice of the hospital. Symptomatic treatment for other chemo/biotherapy-related AEs events are also allowed.

STUDY MEDICATION

Eligible patients with H&N cancer undergoing chemo/bioradiation are assigned at 1:1 ratio to receive:

- Group A: MLT 3% mucoadhesive oral gel (mouthwashes & swallowing, 5-times a day)
- Group B: matching placebo (mouthwashes & swallowing, 5-times a day)

EFFICACY ASSESSMENT

MUCOSITIS GRADE is evaluated using:

- Acute Radiation Morbidity Scoring Criteria from Radiation Therapy Oncology Group (RTOG), **and**
- National Cancer Institute (NCI) Common Terminology Criteria for Adverse Events (CTCAE) version 4.03.

MUCOSITIS grade acc. to the RTOG SCALE					
Morphologic evaluation	0	1	2	3	4
MUCOUS MEMBRANE	No change over baseline	Injection/ may experience mild pain not requiring analgesic	Patchy mucositis which may produce an inflammatory serosanguinitis discharge/ may experience moderate pain requiring analgesia	Confluent fibrinous mucositis/ may include severe pain requiring narcotic	Ulceration, hemorrhage or necrosis
MUCOSITIS grade acc. to the NCI-CTCAE SCALE					
Functional evaluation		1	2	3	4
MUCOSITIS ORAL		Asymptomatic or mild symptoms; intervention not indicated.	Moderate pain; not interfering with oral intake; modified diet indicated.	Severe pain; interfering with oral intake	Life-threatening consequences; urgent intervention indicated

ENDPOINTS

Primary endpoint: Number (percentage) of patients who develop severe OM (≥G3 as per RTOG).

Secondary endpoints:

<u>Efficacy:</u>
Number (%) of patients who develop SOM (G3-G4 acc. to NCI-CTCAE)
Number of days with OM of any grade acc. to the RTOG scale
Number of days with G3-G4 OM acc. to the RTOG scale and
Time to onset of G3-G4 OM acc. to the RTOG scale from starting CT

<u>Safety:</u>
Num(%) of patients with G1-G4 NCI-CTCAE AEs related to IMP
Num(%) of patients who develop CIS or CET-associated G1-G4 AEs (NCI-CTCAE)
Num(%) of patients who develop RT-associated AEs different from OM (RTOG)

<u>Quality of Life:</u>
Change from baseline in EORTC QLQ-C30 and EORTC QLQ-H&N35 scores
Change from baseline in ECOG-Performance status score

<u>Pharmacokinetics:</u> C_{max}, C_{min}, T_{max}, AUC, $T_{1/2}$, V_d and clearance.

Additional endpoints:
Change from baseline in oral pain intensity (VAS at diff. time points)
Num (%) of patients who need minor or major opioids
Num (%) of patients who need special procedures on nutritional status
RT treatment breaks, Total dose and intensity of RT and CT administered (mg/m^2)
Num (%) of patients with CR, PR, SD and PD using the RECIST 1.1 criteria.
Num (%) of patients who develop OM (G1-G2 acc. to the RTOG scale)
Num (%) of patients who develop OM (G1-G2 acc. to NCI-CTCAE)
Num (%) of patients who develop OM (G ≥1 acc. to RTOG scale)
Num (%) of patients who develop OM (G ≥1 acc. to NCI-CTCAE)
Num of days with any grade of OM and with ≥ G3 OM acc. to NCI-CTCAE
Time between the start of the CT until the onset of G3-G4 OM (NCI-CTCAE)
Time between the start of RT until onset of G3-G4 OM (NCI-CTCAE & RTOG)

TUMOUR ASSESSMENT

CT or MRI Imaging for tumour assessment according to RECIST 1.1. criteria (approx. 2 months after the end of RT, following routine clinical practice).

RESULTS

The first patient was enrolled in November 2015 and, up to may 2017, 69 patients were randomized to treatment with either MLT or placebo oral gel. Results are expected by 1Q 2018.

CONCLUSIONS

This prospective, randomized, double-blind and placebo-controlled study will allow to assess whether MLT 3% mucoadhesive oral gel is safe and has been able to prevent severe OM in H&N cancer patients undergoing C/B-RT, and whether if has shown efficacy in the other evaluated endpoints.

MASCC/ISOO 2017 Washington 22-24 June 2017