Presented at Annual Meeting on Supportive Care in Cancer • June 22–24, 2017 • Washington, DC

NOVARTIS

Health-Related Quality of Life of Postmenopausal Women With Hormone Receptor-positive Human Epidermal Growth Factor Receptor 2-negative Advanced Breast Cancer Treated With Ribociclib + Letrozole: Results From MONALEESA-2

Sunil Verma,[1] Joyce O'Shaughnessy,[2] Howard Burris,[3] Mario Campone,[4] Emilio Alba,[5] David Chandiwana,[6] Stephanie Manson,[6] Santosh Sutradhar,[6] Mauricio Monaco,[6] Wolfgang Janni[7]

[1]Tom Baker Cancer Centre, Calgary, Canada; [2]Texas Oncology-Baylor Charles A. Sammons Cancer Center and The US Oncology Network, Dallas, TX; [3]Sarah Cannon Research Institute, Nashville, TN; [4]Institut de Cancérologie de l'Ouest/René Gauducheau, Saint-Herblain, France; [5]Hospital Universitario Virgen de la Victoria, IBIMA, Málaga, Spain; [6]Novartis Pharmaceuticals Corporation, East Hanover, NJ; [7]Universitätsklinikum Ulm, Ulm, Germany

References

1. Hortobagyi GN et al. *N Engl J Med* 2016;375:1738–1748.
2. Finn RS et al. *N Engl J Med* 2016;375:1925–1936.
3. Cristofanilli M et al. *Lancet Oncol* 2016;17:425–439.
4. Butters DJ et al. *Cochrane Database Syst Rev* 2010;CD003368.
5. Miles D et al. *Oncologist* 2002;7 Suppl 6:13–19.
6. Hortobagyi GN et al. ASCO 2017;abst 1038.
7. Aaronson NK et al. *J Natl Cancer Inst* 1993;85:365–376.
8. Sprangers MA et al. *J Clin Oncol* 1996;14:2756–2768.
9. Osoba D et al. *J Clin* Oncol 1998;16:139–144.

Background

The addition of a cyclin-dependent kinase (CDK) 4/6 inhibitor to endocrine therapy improves clinical outcomes compared with endocrine therapy alone in patients with hormone receptor-positive (HR+), human epidermal growth factor receptor 2-negative (HER2–) advanced breast cancer (ABC).[1–3]

- When evaluating new treatments in this setting, it is important to assess the quality of the time gained by delaying disease progression using patient-reported outcomes (PROs) as a component of benefit–risk assessments.
- While delaying disease progression may help maintain patient quality of life (QoL), the addition of novel treatments to existing therapies can add toxicities, which may diminish QoL.[4,5]

In the MONALEESA-2 study, first-line treatment with ribociclib (selective CDK4/6 inhibitor) + letrozole significantly prolonged progression-free survival (PFS) and showed higher overall response rates vs placebo + letrozole in postmenopausal women with HR+, HER2– ABC.[1]

- At the planned interim analysis (cut-off date: January 29, 2016), median PFS in the ribociclib + letrozole arm was not reached vs 14.7 months in the placebo + letrozole arm (hazard ratio=0.556; 95% confidence interval [CI]: 0.429, 0.720; $p=3.29 \times 10^{-6}$).[1]
- Updated PFS analyses (cut-off date: January 2, 2017) demonstrated continued treatment benefit with ribociclib; median PFS was 25.3 months in the ribociclib + letrozole arm vs 16.0 months in the placebo + letrozole arm (hazard ratio=0.568; 95% CI: 0.457, 0.704; $p=9.63 \times 10^{-8}$).[6]

Here we report validated, cancer-specific PROs from the MONALEESA-2 trial to highlight patient experience, focusing on health-related quality of life (HRQoL) and symptoms.

Objectives

Evaluate HRQoL and symptoms with ribociclib + letrozole vs placebo + letrozole as change from baseline, time to definitive 10% deterioration, and mean on-treatment vs end of treatment (EOT) scores in the global health status/QoL scale score of the European Organisation for Research and Treatment of Cancer's cancer questionnaire (EORTC QLQ-C30).

Methods

Study design

In the international, Phase III, randomized, double-blind MONALEESA-2 trial (NCT01958021), postmenopausal women with HR+, HER2– ABC who had not received any prior systemic therapy for advanced disease were randomized 1:1 to receive ribociclib (600 mg/day; 3-weeks-on/1-week-off) + letrozole (2.5 mg/day; continuous) or placebo + letrozole.

PRO assessments and analyses

The EORTC QLQ-C30, version 3.0[7] and breast cancer-specific questionnaire (EORTC QLQ-BR23, version 1.0)[8] were used to explore patient-reported HRQoL, functioning, disease symptoms, and treatment-related side effects.

- EORTC QLQ-C30 and EORTC QLQ-BR23 are recognized reliable and valid measures frequently used in clinical trials of patients with ABC.[7,8]

HRQoL questionnaires were completed by patients at the beginning of each visit at screening, every 8 weeks following randomization for the first 18 months, every 12 weeks thereafter until disease progression, death, loss to follow-up, or withdrawal of consent, and at treatment discontinuation.

Changes from baseline in all subscales were analyzed using a linear effect model that included treatment, stratification factor, and baseline score.

Time to definitive 10% deterioration in HRQoL score from baseline (without improvement at subsequent on-treatment visits) was compared between treatment arms using the stratified log-rank test.

The difference in mean change in HRQoL from baseline to EOT vs baseline to the visit immediately before the EOT cycle was evaluated using a paired t-test.

Results were defined as meaningful:

- Statistically, using appropriate post-hoc tests for statistical significance;
- Clinically, based on a clinically meaningful threshold for change; for EORTC QLQ-C30, the threshold for a minimally important difference (MID) was a change of 5–10 points.[9]

Results

Patients

668 patients were randomized to receive ribociclib + letrozole (n=334) or placebo + letrozole (n=334) between January 2014 and March 2015.

- Patient compliance with completing HRQoL questionnaires was high, with >90% compliance up to Cycle 19. The sample size declined over time as patients did not complete questionnaires following disease progression.

Overall HRQoL

During treatment, overall HRQoL (global health status/QoL score) was maintained from baseline and was similar in both arms (**Figure 1**).

- There were no statistically significant differences in HRQoL between arms during treatment.
- Differences in HRQoL between arms during treatment were less than the MID.

At disease progression/EOT, mean overall HRQoL worsened numerically in both treatment arms compared with mean on-treatment HRQoL (**Table 1**).

Figure 1. Change From Baseline in Global Health Patient-reported Outcomes, by Treatment Arm – Global Health Status/QoL Scale Score of EORTC QLQ-C30

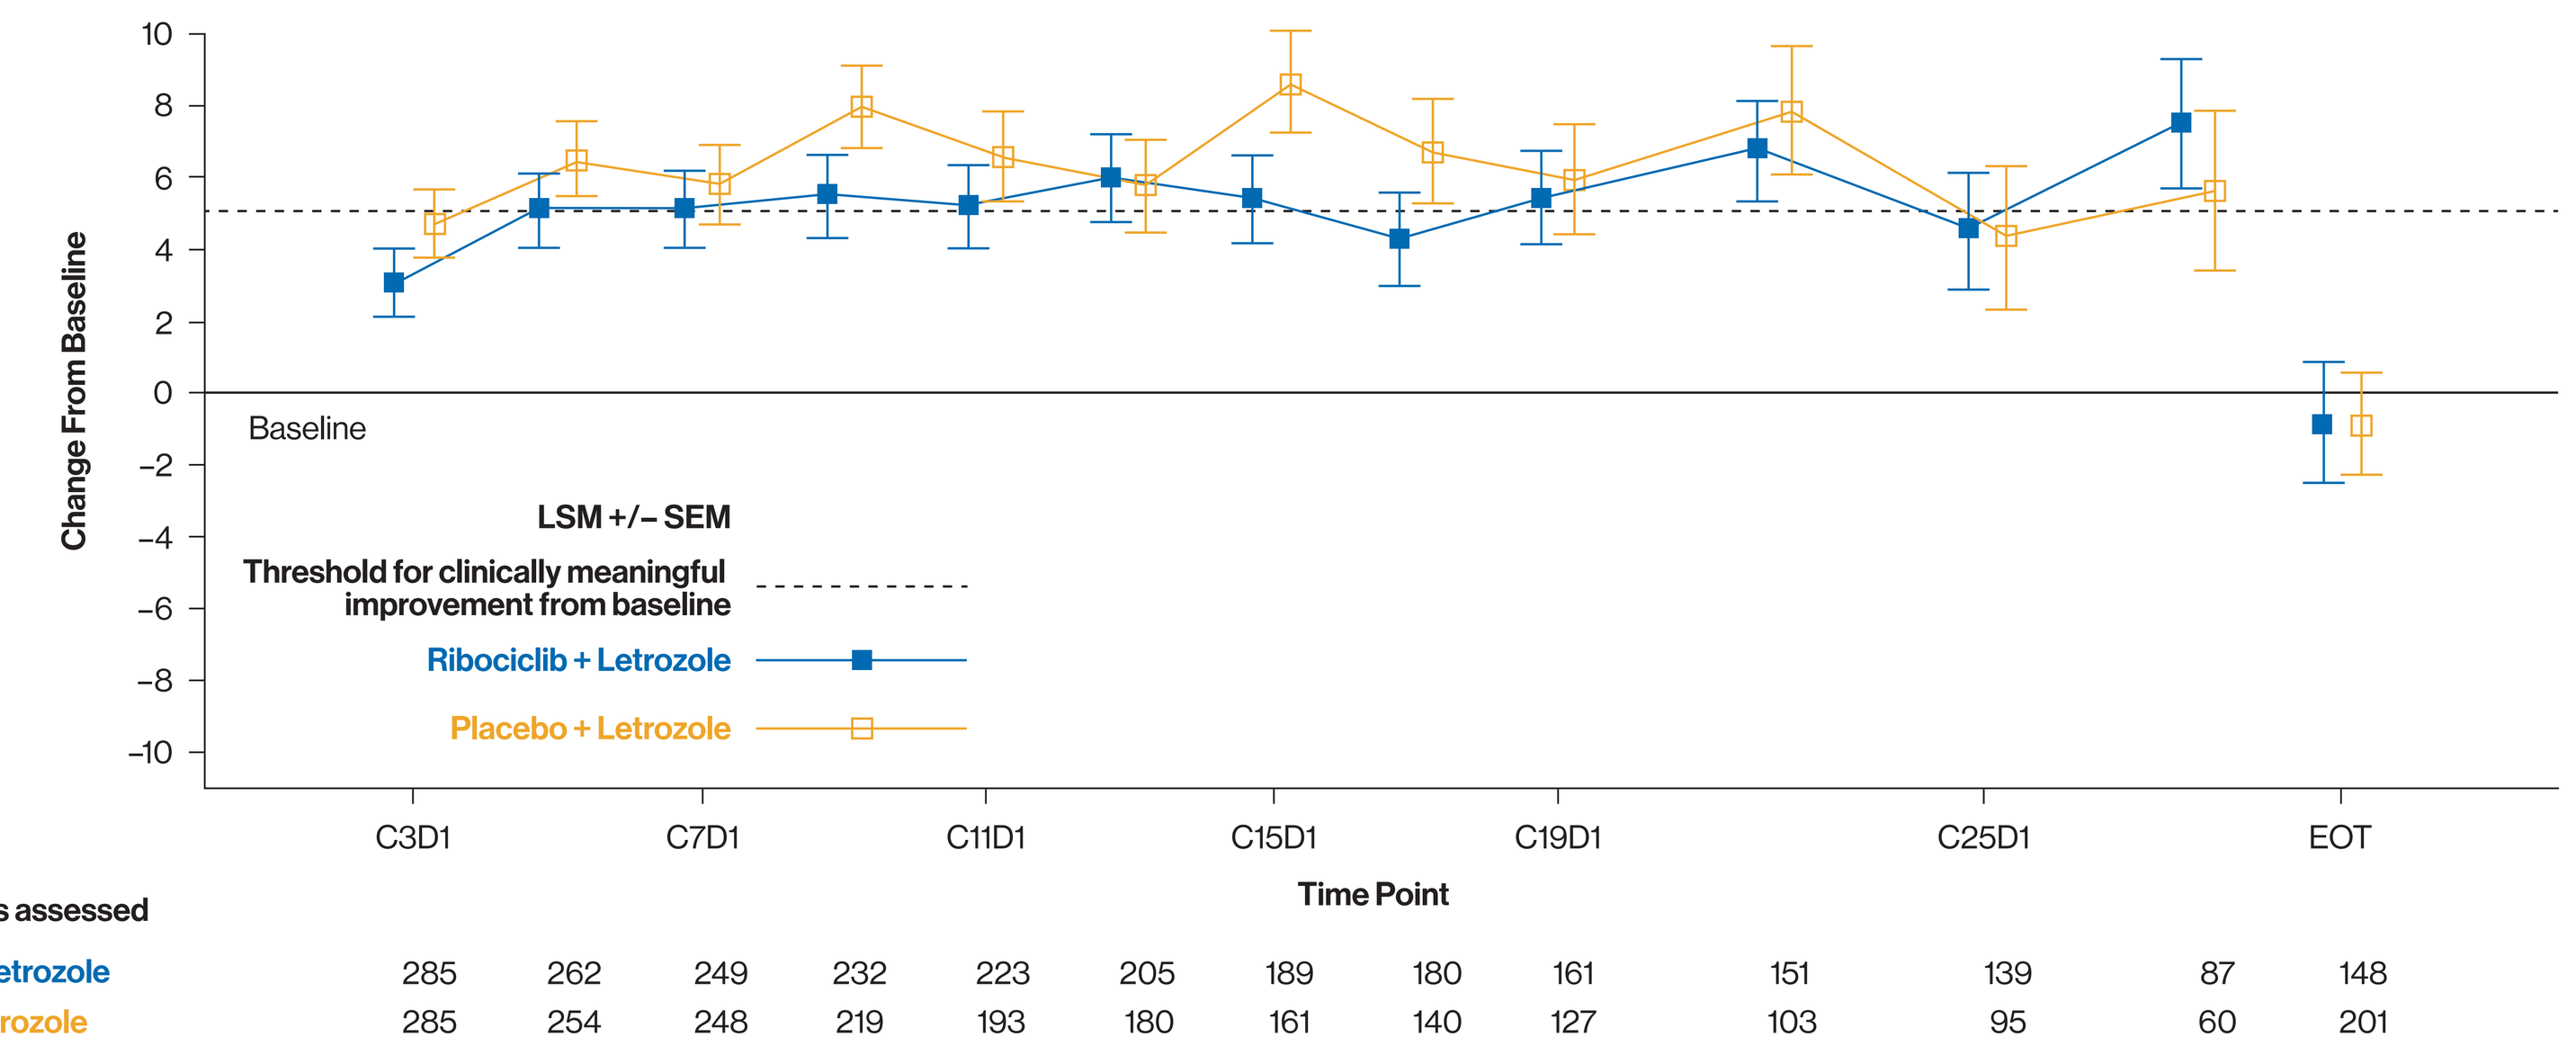

Change From Baseline
10
8
6
4
2
0
−2
−4
−6
−8
−10
Baseline
LSM +/– SEM
Threshold for clinically meaningful improvement from baseline
Ribociclib + Letrozole
Placebo + Letrozole
C3D1
C7D1
C11D1
C15D1
C19D1
C25D1
EOT
Time Point
No. of patients assessed
Ribociclib + Letrozole 285 262 249 232 223 205 189 180 161 151 139 87 148
Placebo + Letrozole 285 254 248 219 193 180 161 140 127 103 95 60 201

C, Cycle; D, Day; EOT, end of treatment; LSM, least squares mean; SEM, standard error of the mean.
Data cut-off: January 4, 2017. The time profile provides the average estimates for the change from baseline for the interval from baseline up to the respective cycle as derived from the linear effects model. Positive changes from baseline are related to improvement in HRQoL. >5 point improvement from baseline in HRQoL score defined as clinically meaningful.
Only patients with baseline scores and at least one non-missing post-baseline assessment are included for change from baseline analysis which was performed using the linear effect model with treatment, stratification factor, and baseline score in the model.

Table 1. Summary of EORTC QLQ-C30 Global Health Status/QoL Scores by Treatment

Change From Baseline, Mean (SD)	Statistic	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334
From Cycle 3 Day 1 to Cycle 27 Day 1	n*	2276	2005
	Mean (SD)	4.6 (21.4)	6.7 (20.1)
	Median (range)	0 (−83, 100)	0 (−67, 67)
EOT	n*	148	201
	Mean (SD)	−0.5 (22.6)	−1.2 (24.5)
	Median (range)	0 (−58, 67)	0 (−100, 50)

EOT, end of treatment; SD, standard deviation.
*n is the number of observations; a patient may have more than one observation.
Data cut-off: January 4, 2017. Analysis only includes assessments up to the time point (Cycle 27 Day 1) where there are at least 50 patients on each of the treatments. 5–10-point change in HRQoL score defined as clinically meaningful.

Overall HRQoL worsened numerically in both treatment arms at EOT compared with on-treatment HRQoL immediately before EOT, with noticeable worsening in patients receiving placebo + letrozole.

- The difference in mean overall HRQoL scores between the visit immediately before the EOT cycle, and EOT, was 2.9 (95% CI: −0.1, 5.9) in the ribociclib + letrozole arm and 4.7 (95% CI: −1.9, 7.6) in the placebo + letrozole arm (**Table 2**).

Table 2. Difference in EORTC QLQ-C30 Global Health Status/QoL Scores Immediately Before EOT and at EOT by Treatment

Change From Baseline, Mean (SD)	Statistic	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334
Visit immediately before EOT cycle (A)	n	135	174
	Mean (SD)	2.3 (21.4)	3.3 (20.8)
	Median (range)	0 (−58, 50)	0 (−67, 50)
EOT (B)	n	135	174
	Mean (SD)	−0.6 (22.4)	−1.5 (24.9)
	Median (range)	0 (−58, 50)	0 (−100, 50)
Difference (A–B)	n	135	174
	Mean (95% CI)	2.9 (−0.1, 5.9)	4.7 (1.9, 7.6)
	p-value	0.059	0.001

CI, confidence interval; EOT, end of treatment; SD, standard deviation.
Data cut-off: January 4, 2017. Analysis only includes patients with EOT assessments.
p-value is calculated based on a paired t-test. 5–10-point change in HRQoL score defined as clinically meaningful.

Time to definitive deterioration of the global health status/QoL scale score of the EORTC QLQ-C30 questionnaire by at least 10% was similar between treatment arms (hazard ratio=0.944; 95% CI: 0.720, 1.237; **Figure 2**).

Ribociclib was discovered by Novartis Institutes for BioMedical Research in collaboration with Astex Pharmaceuticals.
This study was sponsored by Novartis Pharmaceuticals Corporation.

Figure 2. Time to Definitive Deterioration of Global Health Status/QoL Scale Score of EORTC QLQ-C30 From Baseline by At Least 10%

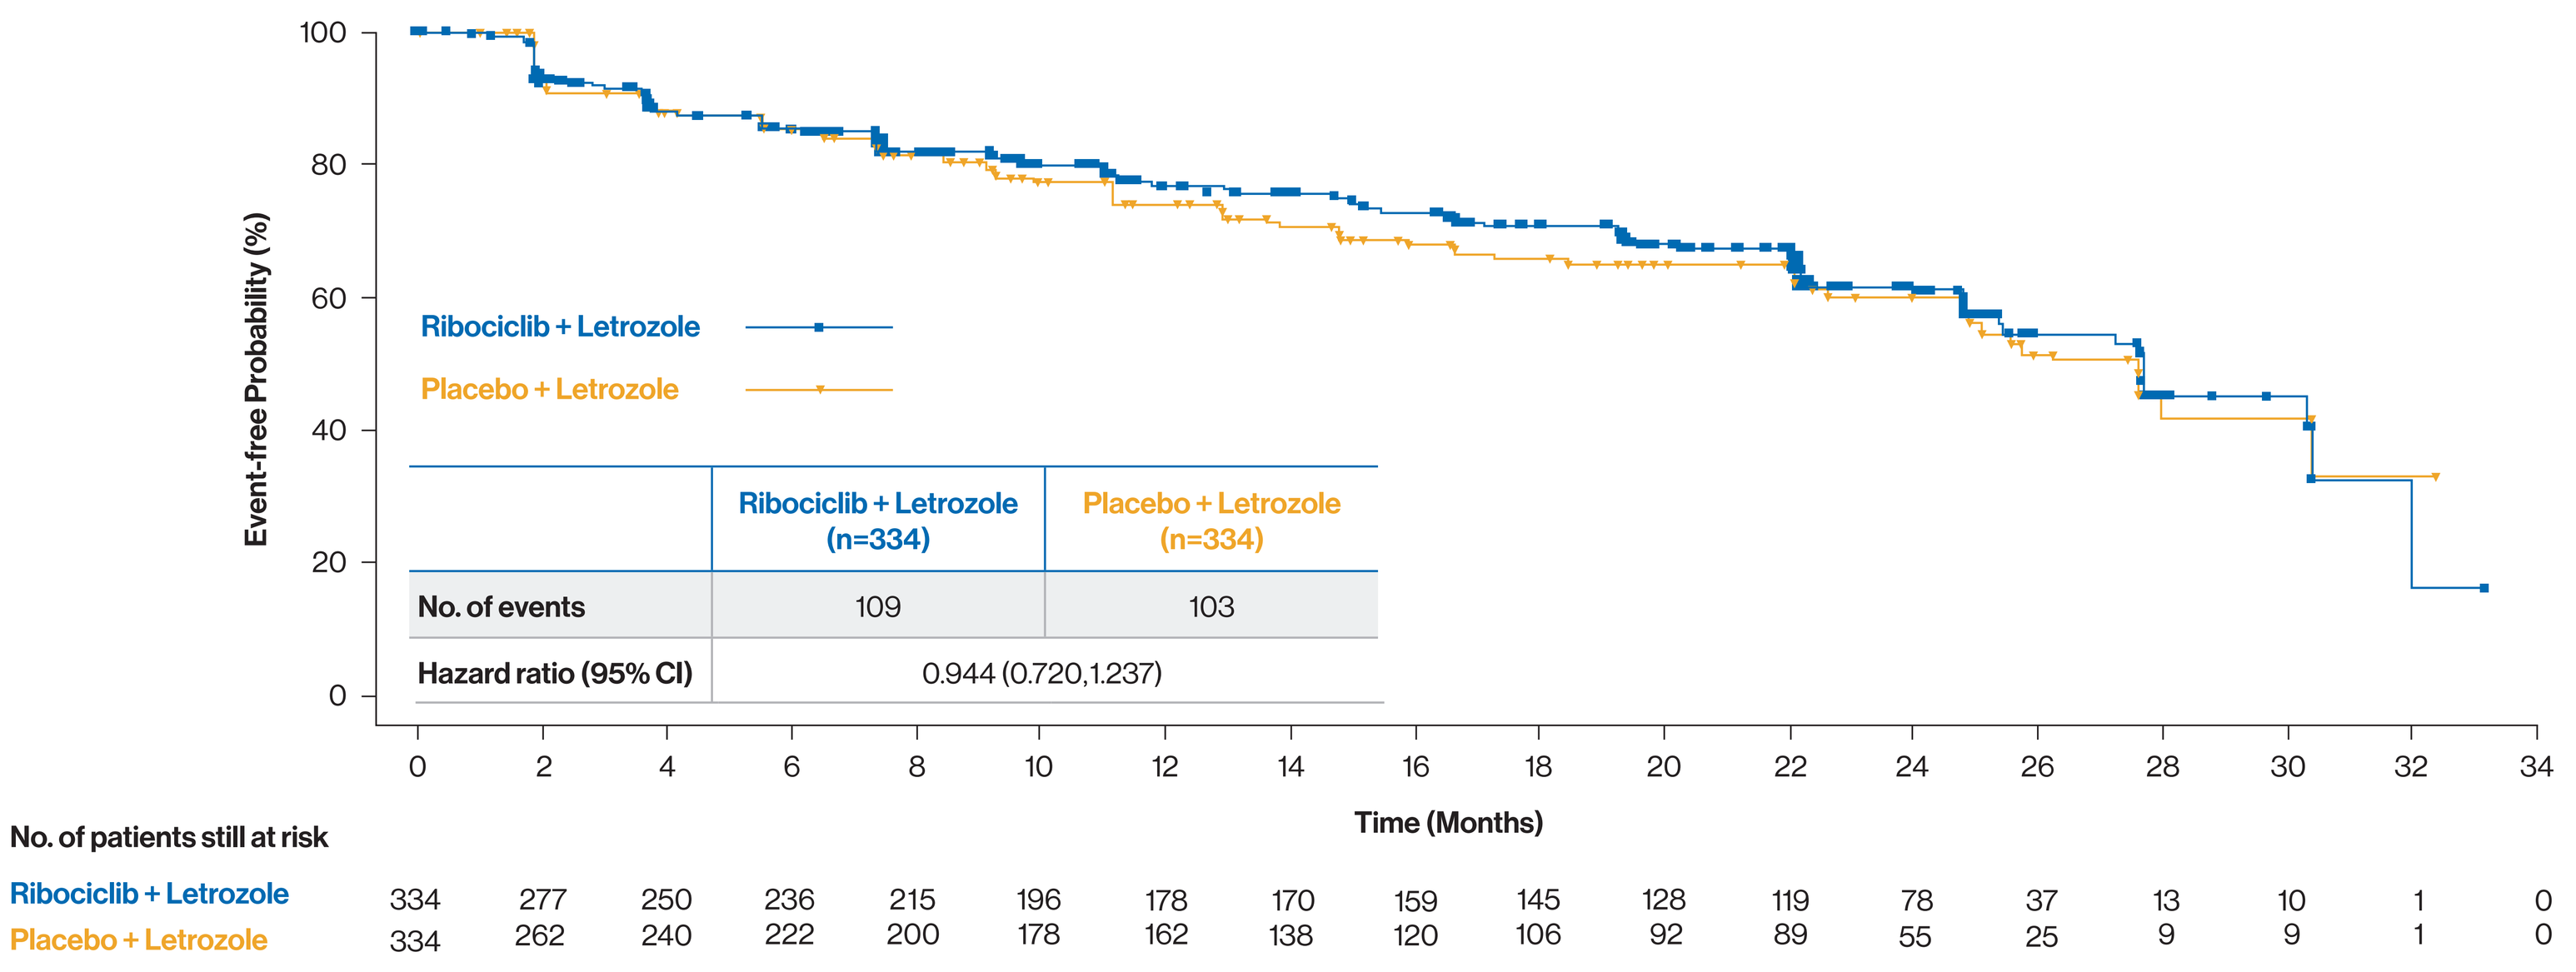

Event-free Probability (%)
100
80
60
40
20
0
Ribociclib + Letrozole
Placebo + Letrozole
Ribociclib + Letrozole (n=334)
Placebo + Letrozole (n=334)
No. of events 109 103
Hazard ratio (95% CI) 0.944 (0.720,1.237)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34
Time (Months)
No. of patients still at risk
Ribociclib + Letrozole 334 277 250 236 215 196 178 170 159 145 128 119 78 37 13 10 1 0
Placebo + Letrozole 334 262 240 222 200 178 162 138 120 106 92 89 55 25 9 9 1 0

CI, confidence interval.
Data cut-off: January 4, 2017. Full analysis set.

Key symptoms

No statistically or clinically meaningful differences were observed between the two arms for key symptoms using the EORTC QLQ-C30 questionnaire, including fatigue, nausea, and vomiting (**Tables 3** and **4**).

- Symptom scores were generally higher in the ribociclib + letrozole vs placebo + letrozole arm, but these differences were not considered clinically meaningful (i.e. did not exceed the MID).
- At an earlier data cut-off (January 29, 2016), similar results were observed for additional EORTC QLQ-C30 questionnaire domains, including physical, emotional, cognitive, and social functioning (data not shown) and for EORTC QLQ-BR23 questionnaire domains, including future perspective, side effects, and upset by hair loss (**Table 5**).

Table 3. Nausea and Vomiting Scale Score of EORTC QLQ-C30 – Mean Change From Baseline by Treatment and Visit

Change From Baseline, Mean (SD)	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334	Treatment Difference (Ribociclib–Placebo), Mean (95% CI)
Cycle 3 Day 1, n=569	2.9 (16.9)	−0.2 (16.3)	3.1 (0.4, 5.9)
Cycle 5 Day 1, n=519	1.9 (18.3)	−2.6 (17.6)	4.5 (1.4, 7.6)
Cycle 7 Day 1, n=499	0.1 (17.7)	−3.9 (17.7)	4.1 (1.0, 7.2)
Cycle 9 Day 1, n=451	−0.8 (15.9)	−2.7 (18.5)	1.9 (−1.3, 5.1)
Cycle 11 Day 1, n=418	1.0 (18.4)	−2.3 (19.5)	3.4 (−0.3, 7.0)
Cycle 13 Day 1, n=388	−1.1 (15.3)	−2.2 (21.0)	1.0 (−2.6, 4.7)
Cycle 15 Day 1, n=352	0.3 (16.4)	−3.3 (16.1)	3.5 (0.1, 7.0)
Cycle 17 Day 1, n=321	0.5 (17.9)	−1.8 (15.3)	2.2 (−1.5, 6.0)
Cycle 19 Day 1, n=289	0 (16.1)	−2.9 (12.3)	2.9 (−0.5, 6.3)
Cycle 22 Day 1, n=251	−0.3 (16.7)	−3.1 (14.7)	2.7 (−1.3, 6.8)
Cycle 25 Day 1, n=235	0.5 (16.9)	−0.7 (14.5)	1.2 (−3.0, 5.3)
Cycle 28 Day 1, n=146	0.4 (17.6)	−0.8 (14.7)	1.2 (−4.3, 6.7)
EOT, n=350	4.4 (19.4)	0.4 (21.5)	4.0 (−0.4, 8.4)

CI, confidence interval; EOT, end of treatment; SD, standard deviation.
Data cut-off: January 4, 2017. Only patients with baseline scores and at least one non-missing post-baseline assessment were included in the analysis.
Only time points with data available for at least 50 patients in each treatment arm are included. 5–10-point change or difference in score defined as clinically meaningful.

Table 4. Fatigue Scale Score of EORTC QLQ-C30 – Mean Change From Baseline by Treatment and Visit

Change From Baseline, Mean (SD)	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334	Treatment Difference (Ribociclib–Placebo), Mean (95% CI)
Cycle 3 Day 1, n=572	2.0 (20.5)	−0.2 (20.8)	2.2 (−1.2, 5.6)
Cycle 5 Day 1, n=524	−1.1 (20.0)	−3.1 (20.8)	2.1 (−1.4, 5.6)
Cycle 7 Day 1, n=501	−0.9 (19.9)	−3.4 (21.3)	2.6 (−1.0, 6.2)
Cycle 9 Day 1, n=453	−0.2 (20.8)	−2.8 (21.9)	2.6 (−1.4, 6.5)
Cycle 11 Day 1, n=422	0.5 (21.9)	−2.2 (22.3)	2.7 (−1.5, 6.9)
Cycle 13 Day 1, n=390	−0.5 (21.6)	−3.2 (22.1)	2.7 (−1.7, 7.0)
Cycle 15 Day 1, n=353	0.2 (23.8)	−4.4 (22.1)	4.6 (−0.2, 9.5)
Cycle 17 Day 1, n=323	−1.3 (21.4)	−2.8 (22.4)	1.5 (−3.3, 6.3)
Cycle 19 Day 1, n=290	−0.4 (20.4)	−3.0 (21.5)	2.7 (−2.2, 7.5)
Cycle 22 Day 1, n=255	1.0 (23.7)	−4.6 (19.8)	5.6 (0.0, 11.2)
Cycle 25 Day 1, n=235	2.0 (23.6)	−0.7 (22.4)	2.7 (−3.4, 8.7)
Cycle 28 Day 1, n=147	2.8 (22.2)	−0.7 (20.8)	3.6 (−3.6, 10.7)
EOT, n=351	4.7 (23.4)	3.5 (26.4)	1.3 (−4.1, 6.6)

CI, confidence interval; EOT, end of treatment; SD, standard deviation.
Data cut-off: January 4, 2017. Only patients with baseline scores and at least one non-missing post-baseline assessment were included in the analysis.
Only time points with data available for at least 50 patients in each treatment arm are included. 5–10-point change or difference in score defined as clinically meaningful.

Table 5. Future Perspective, Side Effects, and Upset by Hair Loss Scores of EORTC QLQ-BR23 – Mean Score by Treatment and Visit

EORTC QLQ-BR23 Mean Score	Future Perspective		Side Effects		Upset by Hair Loss	
	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334	Ribociclib + Letrozole N=334	Placebo + Letrozole N=334
Baseline	41.2	42.2	14.6	15.2	15.4	19.2
Cycle 3 Day 1	49.2	51.6	21.3	17.7	27.7	30.2
Cycle 5 Day 1	54.0	55.4	20.8	17.8	34.1	29.6
Cycle 7 Day 1	53.6	57.1	20.7	17.5	37.5	33.3
Cycle 9 Day 1	56.2	59.7	21.2	17.2	39.5	35.6
EOT	40.5	45.8	24.2	19.8	39.6	28.7
Scale	0 = worst, 100 = best; 5–10-point change defined as clinically meaningful		0 = best, 100 = worst; 5–10-point change defined as clinically meaningful			

EORTC QLQ-BR23, European Organisation for Research and Treatment of Cancer's breast cancer-specific questionnaire; EOT, end of treatment.
Data cut-off: January 29, 2016. Only patients with baseline scores and at least one non-missing post-baseline assessment were included in the analysis.
Only time points with data available for at least 50 patients in each treatment arm are included.

A clinically meaningful (>5 points) improvement from baseline in pain score was maintained up to and including Cycle 15 in the ribociclib + letrozole arm. A mild improvement (≤5 points) was observed in the ribociclib + letrozole arm from Cycle 17 through Cycle 28 (**Figure 3**).

- In general, mild improvement (≤5 points) was observed in the placebo + letrozole arm over the same time period, except for Cycles 7 and 15.

Pain scores increased to slightly above baseline levels at time of disease progression/EOT in both treatment arms.

Figure 3. Change From Baseline in Pain Score of EORTC QLQ-C30

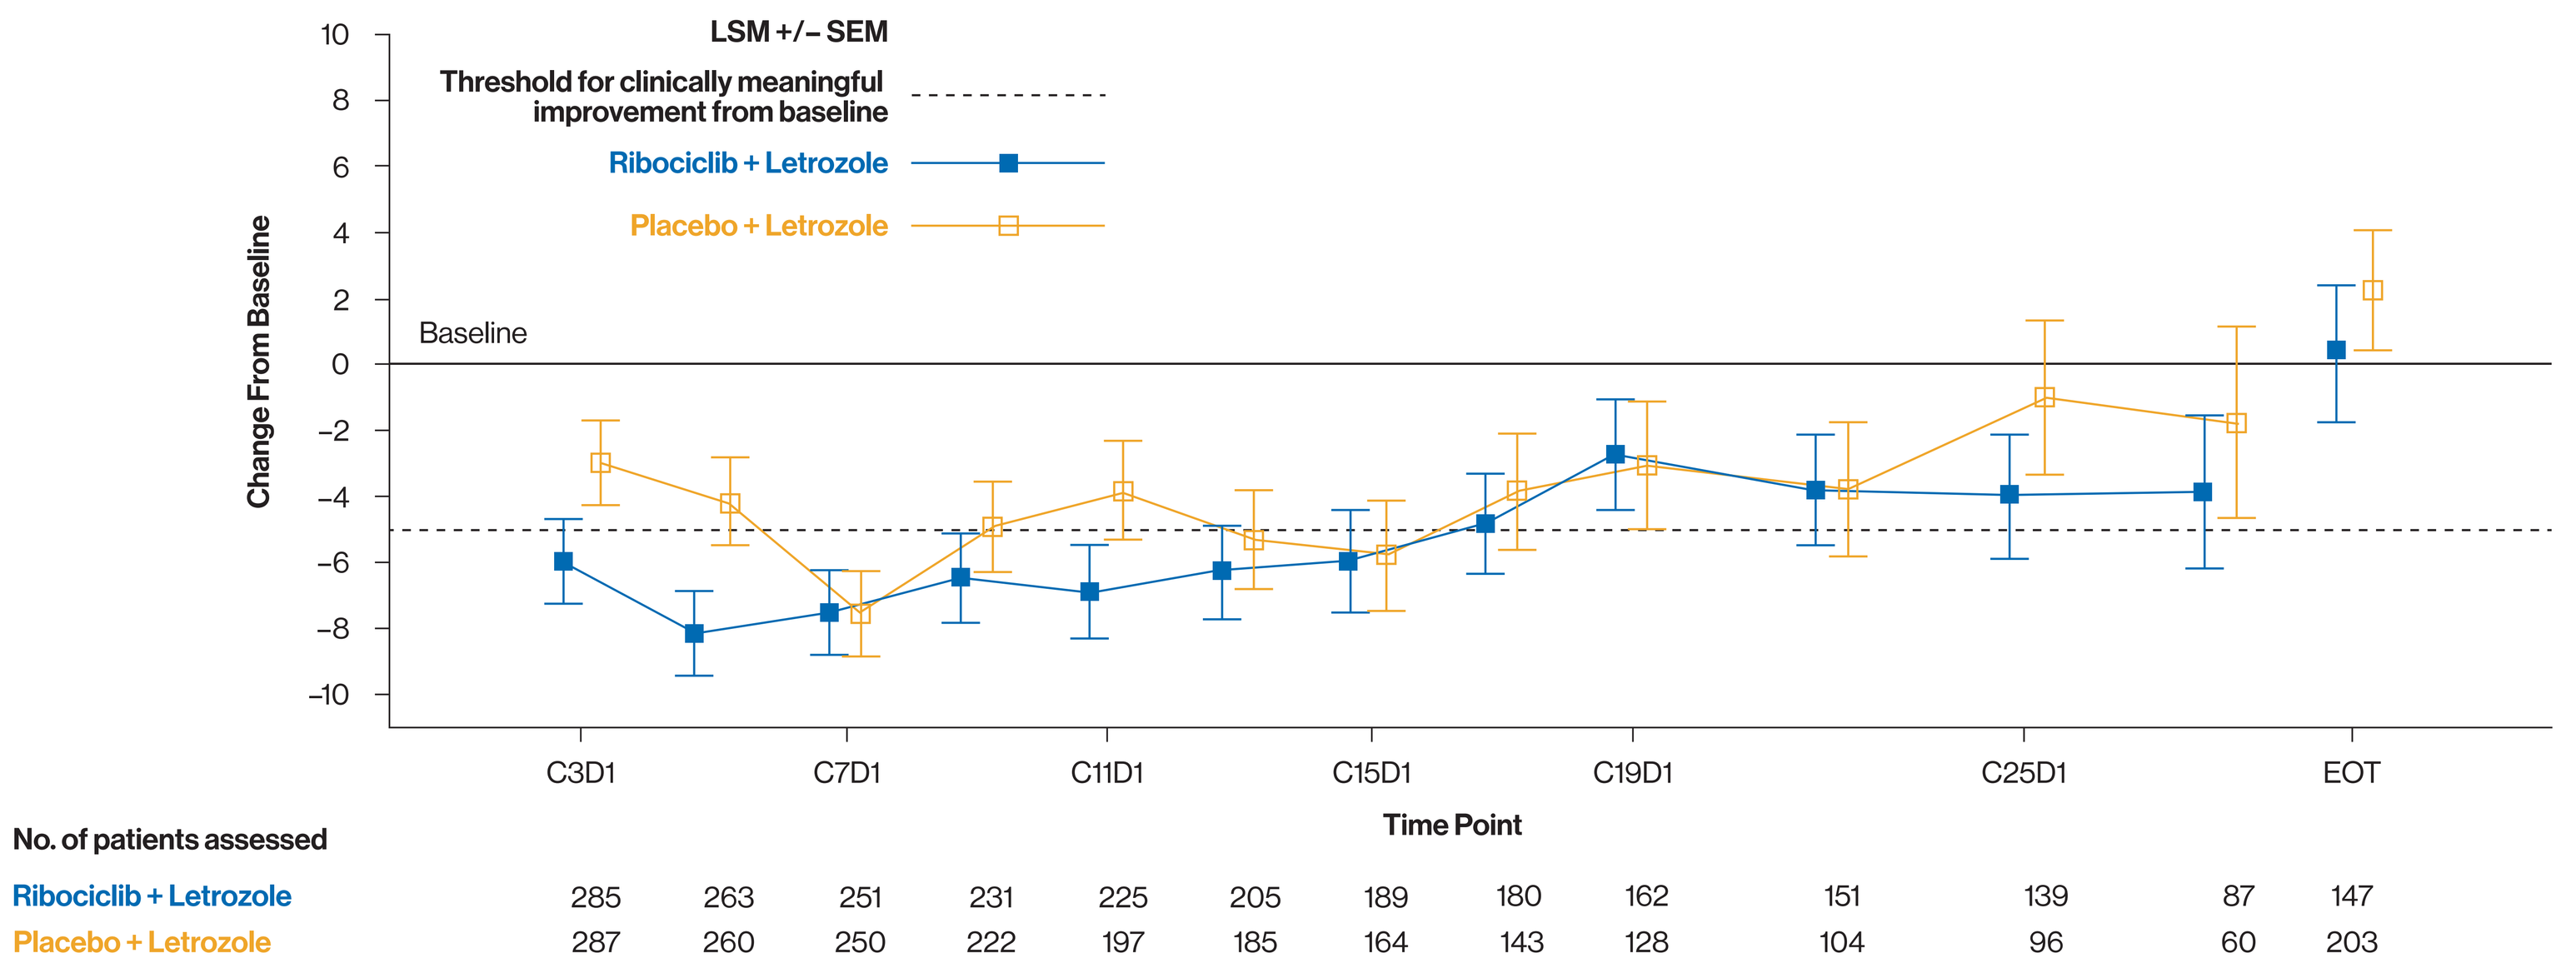

Change From Baseline
10
8
6
4
2
0
−2
−4
−6
−8
−10
LSM +/– SEM
Threshold for clinically meaningful improvement from baseline
Ribociclib + Letrozole
Placebo + Letrozole
Baseline
C3D1
C7D1
C11D1
C15D1
C19D1
C25D1
EOT
Time Point
No. of patients assessed
Ribociclib + Letrozole 285 263 251 231 225 205 189 180 162 151 139 87 147
Placebo + Letrozole 287 260 250 222 197 185 164 143 128 104 96 60 203

C, Cycle; D, Day; EOT, end of treatment; LSM, least squares mean; SEM, standard error of the mean.
Data cut-off: January 4, 2017. The time profile provides the average estimates for the change from baseline to the respective cycle as derived from the linear effects model. Negative changes from baseline are related to reduction in pain. >5 point improvement from baseline in pain score defined as clinically meaningful.
Only patients with baseline scores and at least one non-missing post-baseline assessment are included for change from baseline analysis which was performed using the linear effect model with treatment, stratification factor, and baseline score in the model.

Conclusions

In the MONALEESA-2 study, there were no clinically meaningful differences between treatment arms in HRQoL (i.e. no HRQoL score differences >5 points), suggesting that adverse events did not significantly impact overall HRQoL.

- No statistically significant differences in average HRQoL were observed between treatment arms during treatment (based on post-hoc statistical analyses).
- Clinically meaningful differences between mean on-treatment vs EOT HRQoL scores were observed in both treatment arms.
- Patient-reported symptom scores did not show clinically meaningful differences between treatment arms.
- A clinically meaningful reduction in pain was observed with ribociclib + letrozole treatment, with a non-meaningful trend in pain reduction observed in the placebo + letrozole arm.

In addition to significantly improving PFS compared with placebo + letrozole, ribociclib + letrozole maintains HRQoL in postmenopausal women with HR+, HER2– ABC.

Acknowledgments

The authors would like to thank the patients enrolled in this study, and their families. Editorial assistance was provided by Jennie Pollard MSc and was funded by Novartis Pharmaceuticals Corporation.

Data included in this poster have been previously presented in full at American Society of Clinical Oncology Annual Meeting; June 2–6, 2017; Chicago, IL.

Text: Q9518b
To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=9518b

Scan this QR code
Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
Stephanie.Manson@memorialhermann.org