GENERAL PRACTICE OPTIMISING STRUCTURED MONITORING TO IMPROVE CLINICAL OUTCOMES IN TYPE 2 DIABETES: THE GP-OSMOTIC STUDY PROTOCOL

John Furler[1], David O'Neal[1], Jane Speight[2,3], Irene Blackberry[4], Jo-Anne Manski-Nankervis[1], Jessica Browne[2,3], Kamlesh Khunti[5], Kim Dalziel[1], James Best[6]

[1] University of Melbourne, [2] Deakin University, [3] Australian Centre for Behavioural Research in Diabetes, Diabetes Victoria [4] Latrobe University, [5] University of Leicester, [6] Nanyang Technological University

Introduction

Achieving glycaemic targets is associated with long-term micro and macrovascular benefits. Achieving glycaemic targets in primary care, where the majority of people with type 2 diabetes (T2D) receive their care, is challenging.

Collecting meaningful blood glucose data to guide rational lifestyle and therapy changes to achieve glycaemic targets has been shown to be effective[1] but raises a number of challenges.

New technologies of continuous glucose monitoring offer the opportunity to overcome barriers to collecting meaningful real-time glucose data in a real-world clinical setting. Evidence of their effectiveness in primary care and in people with T2D is limited[2].

Our aim is to assess the (cost) effectiveness of retrospective continuous glucose monitoring (rCGM) in an inclusive population of adults with T2D in primary care. A further aim is to gather robust hypoglycaemia prevalence and glycaemic variability data in this population.

Materials and Methods I

GP-OSMOTIC is an individually randomised controlled trial set in General Practices in Victoria, Australia, testing the effect of the FreeStyle Libre Pro® Flash Glucose Monitoring System (Abbott) applied for 14 days, on 4 occasions per year, compared with usual care.

A collaborative educational consult with a Credentialed Diabetes Educator – Registered Nurse (CDE-RN) will be provided to all participants.

Eligible patients: T2D (> 12 months), aged 18-80 years, diabetes duration >1 - <20 years and most recent HbA1c (in the previous 6 months) >7mmol/mol (0.5%) above their individualised target on maximum oral therapy and/or injectable therapy (insulin and/or GLP1 agonists).

All participating health professionals will take part in a 2 hour training session, with a focus on how to interpret the Ambulatory Glucose Profile (AGP) reports generated by the device. No clinical guidance will be given as patients will be managed according to standard clinical practice.

Materials and methods II

Evaluation at baseline and 12 months will include

- HbA1c (primary outcome), measured as absolute difference in mean change in HbA1c at 12 months between the intervention and control arm;
- time in target (4-10mmol/L), assessed from data downloaded from the rCGM device worn at baseline and 12 months (blind to the control group participants); and
- diabetes distress (Problem Areas in Diabetes (PAID) scale[3]).

We will also measure hypoglycaemia, glycaemic variability, additional patient-reported outcomes, acceptability to health professionals and patients, and cost-effectiveness (informed by EQ5D, the resources used to administer the intervention and health service utilisation data).

Qualitative exploration of experience with and perceived impact of r-CGM and the collaborative care decision-making model will be explored in interviews with a sample of GPs/PNs (~N=10) and intervention participants (~N=20).

Sample size: 50 clinics with 6 participants per clinic (150 in each arm), allowing for 10% clinic attrition and 20% patient attrition giving 80% power to detect a difference in mean HbA1c of 0.5% with a standard deviation of 1.3 and an alpha of 0.05.

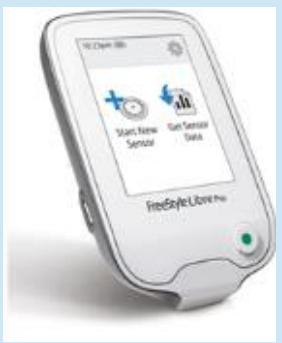

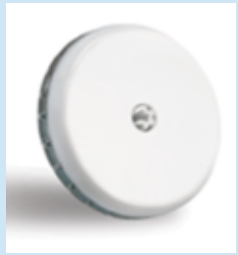

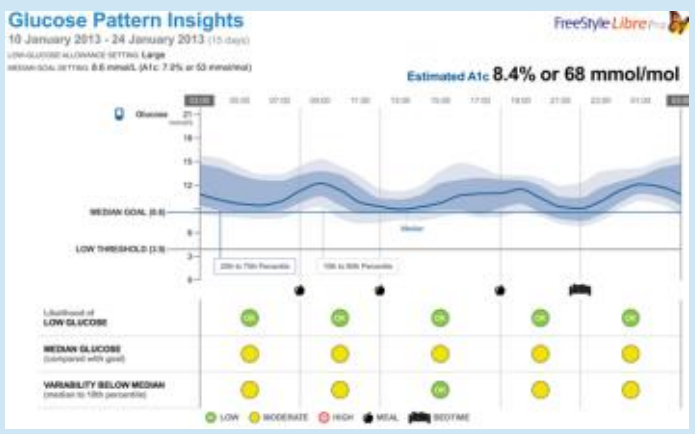
Glucose Pattern Insights
FreeStyle Libre
Estimated A1c 8.4% or 68 mmol/mol

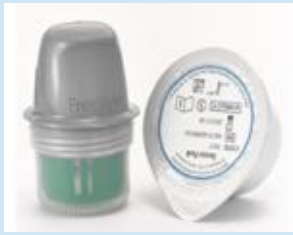

FreeStyle Libre Pro® Flash Glucose Monitoring System Reader, Sensor, applicator pack, and example of AGP report

Conclusions

The OSMOTIC Study will generate cost effectiveness evidence about the potential of Flash rCGM technology to drive personalised treatment intensification to achieve glycaemic targets, with important implications for clinical practice and health policy.

References

1. Polonsky, W. H., et al. (2011). Diabetes Technology & Therapeutics 13(8): 797-802.; 2. Haa et al Diabetes therapy: research, treatment and education of diabetes and related disorders. 2016. Epub 2016/12/22. doi: 10.1007/s13300-016-0223-6. 3. Welch, G. W., et al. (1997). Diabetes Care 20(5): 760-766

Acknowledgments

OSMOTIC is an investigator initiated study, funded by the National Health and Medical Research Council of Australia (NHMRC ID: 1104241). Flash Libre Pro sensors and readers are supplied by Abbott Diabetes Care. Additional funding is provided by Sanofi Australia; **Author contact j.furler@unimelb.edu.au**