A Bioartificial Pancreas for the Treatment of Type 1 Diabetes

U. Barkai[1], Y. Avni[1], B. Zimerman[1], S. Maimon[1], T. Goldman[1], P.de Vos,[2] and A. Rotem[1]

[1]Beta-O_2 Technologies, Rosh-Ha'ain, Israel;
[2]Department of Pathology and Laboratory Medicine, Section of Immunoendocrinology, University Medical Center Groningen, Groningen, The Netherlands.

Introduction

Islet transplantation is only a partial solution for patients with type 1 diabetes (T1D) due to:

- The need for harmful immunosuppressive therapy
- Loss of islets due to limiting amounts of O_2, as the blood supply is disrupted during their isolation

The βAir device

We have developed a bioartificial pancreas, the βAir device, which supplies continuous O_2 and shields the islets from the host immune system, thereby eliminating the need for immunosuppressive drug therapy.[1,2] The device includes (Figures 1,2):

- Biopore porous membranes
- Islets immobilized in hydrogel
- O_2 chamber (which is replenished through the access ports; in the current device, O_2 replenishing is performed every 24 hours)

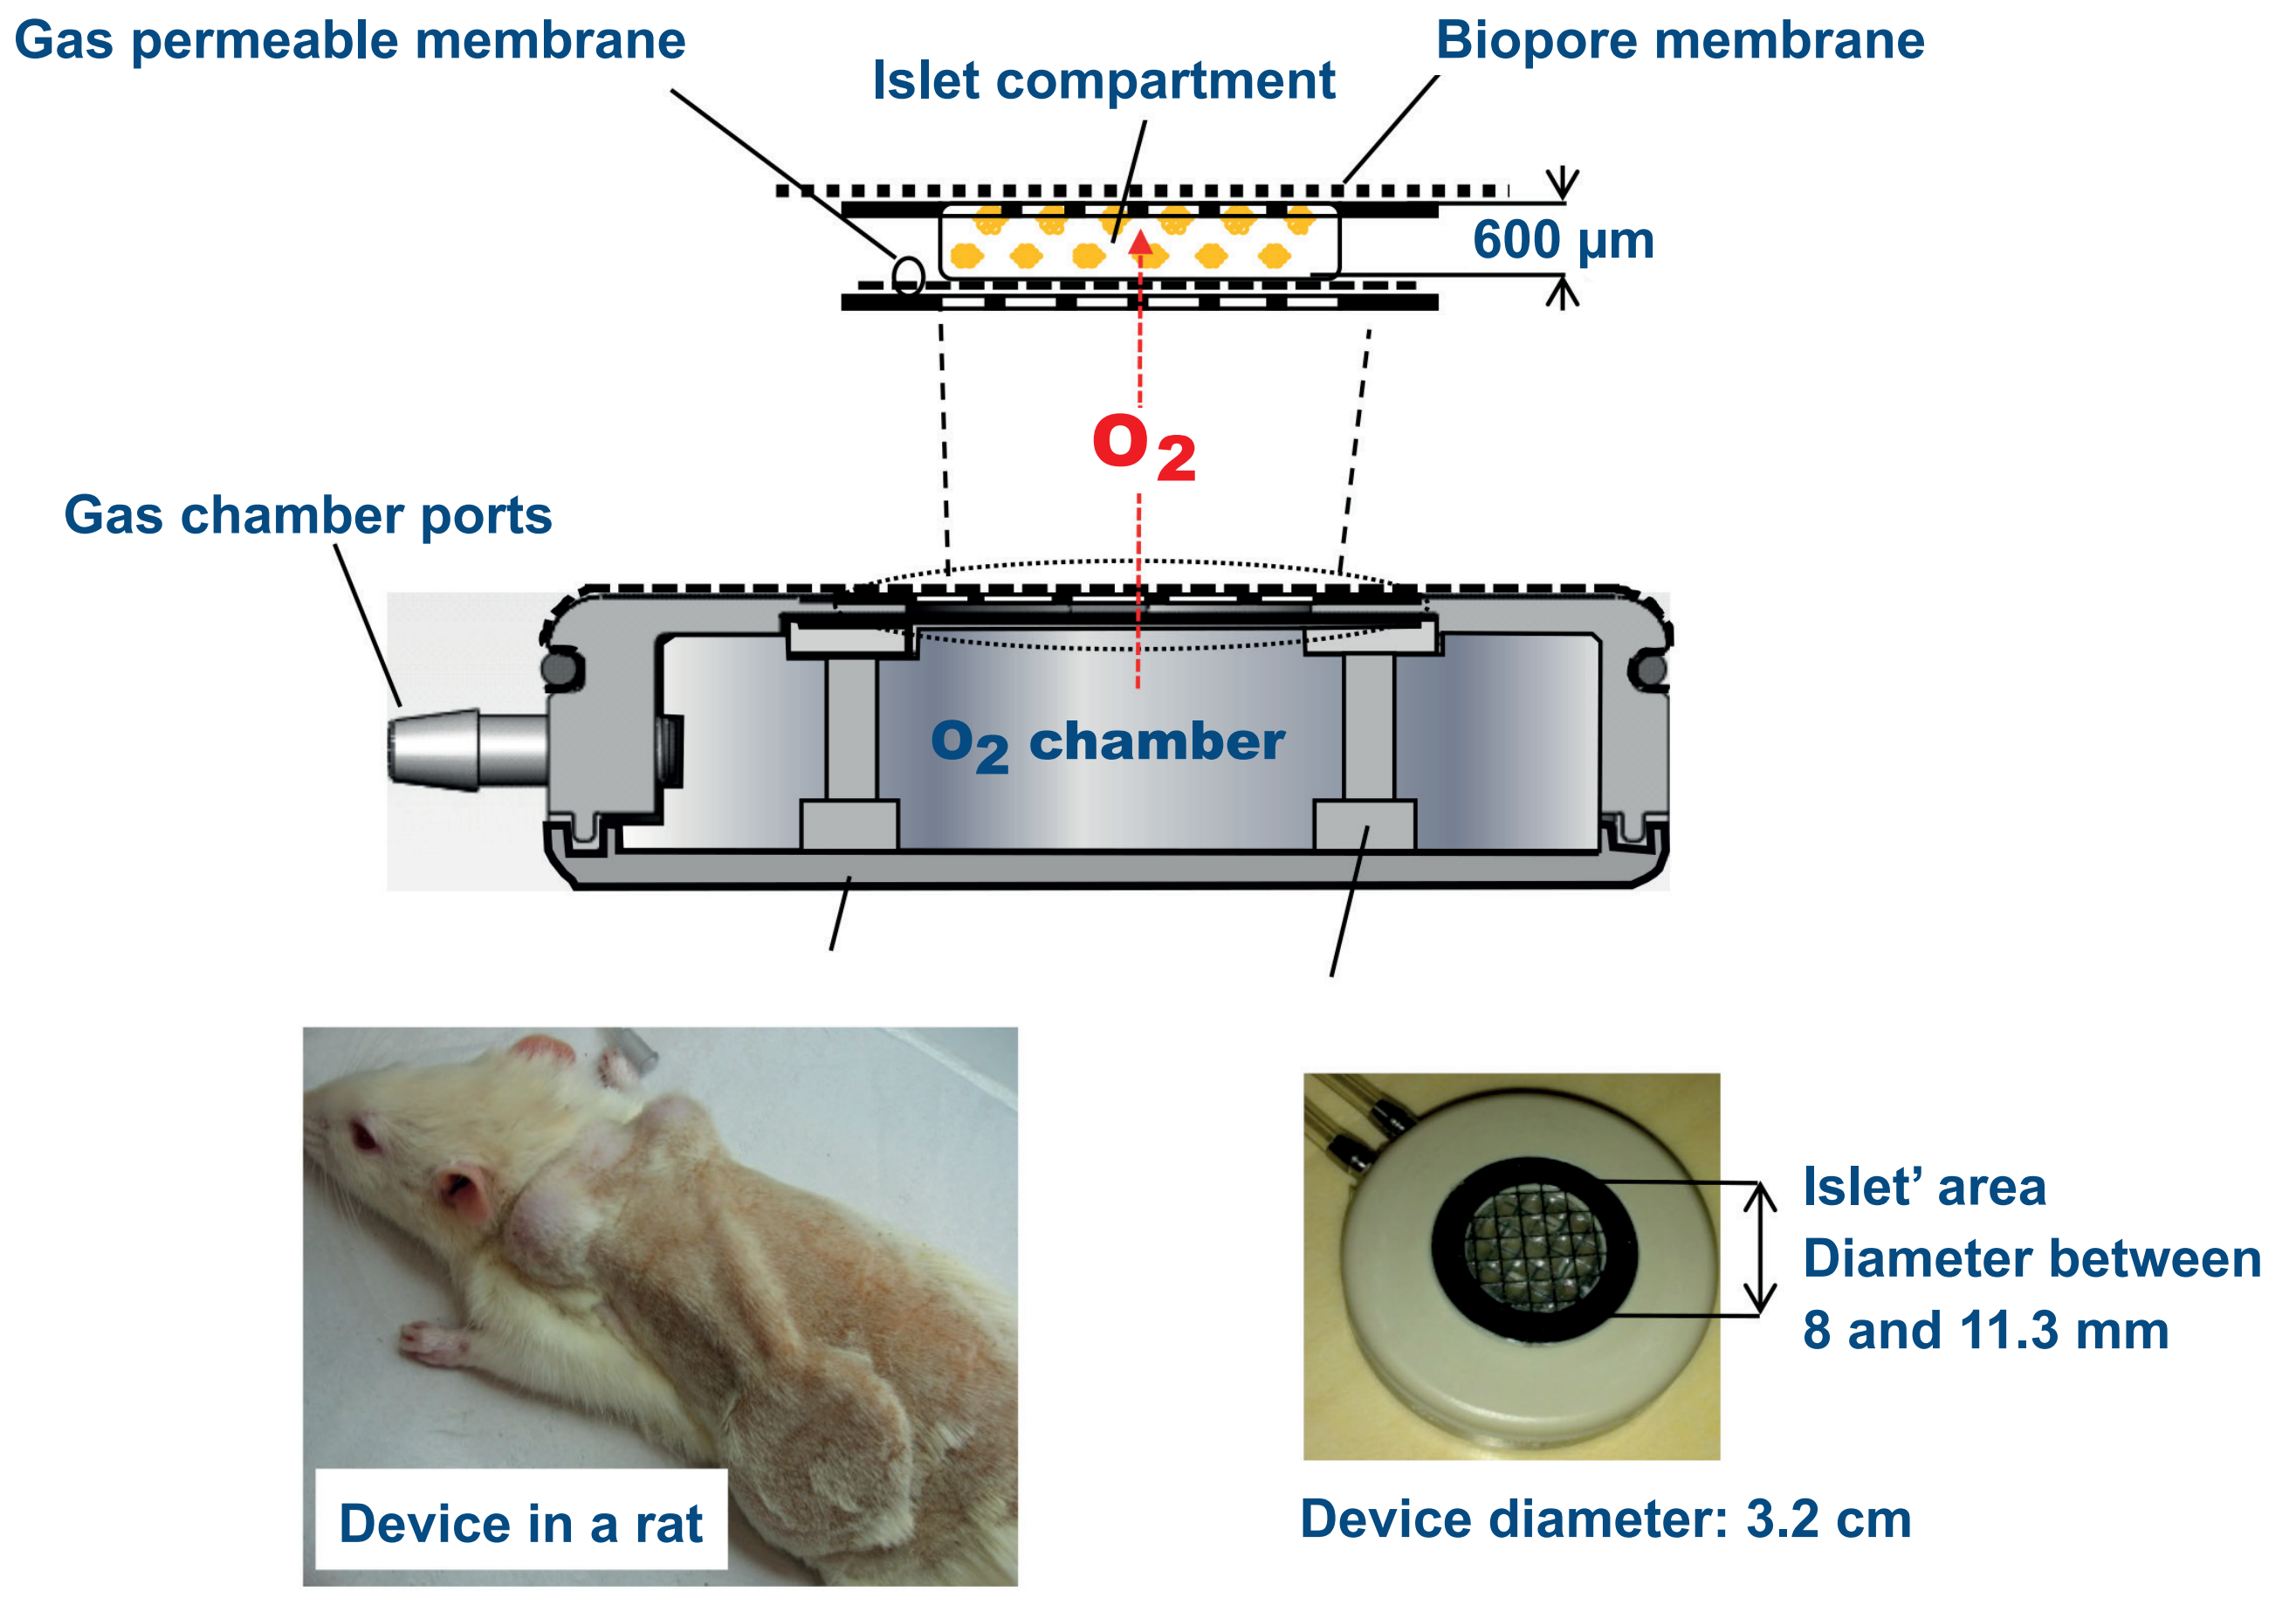

Gas permeable membrane
Islet compartment
Biopore membrane
600 µm
O2
Gas chamber ports
O2 chamber
Device in a rat
Islet' area
Diameter between
8 and 11.3 mm
Device diameter: 3.2 cm

Figure 1. Rat βAir device. Top: Schematic illustration. Bottom left: Device implanted subcutaneously in a rat. Bottom right: Top view of a device.

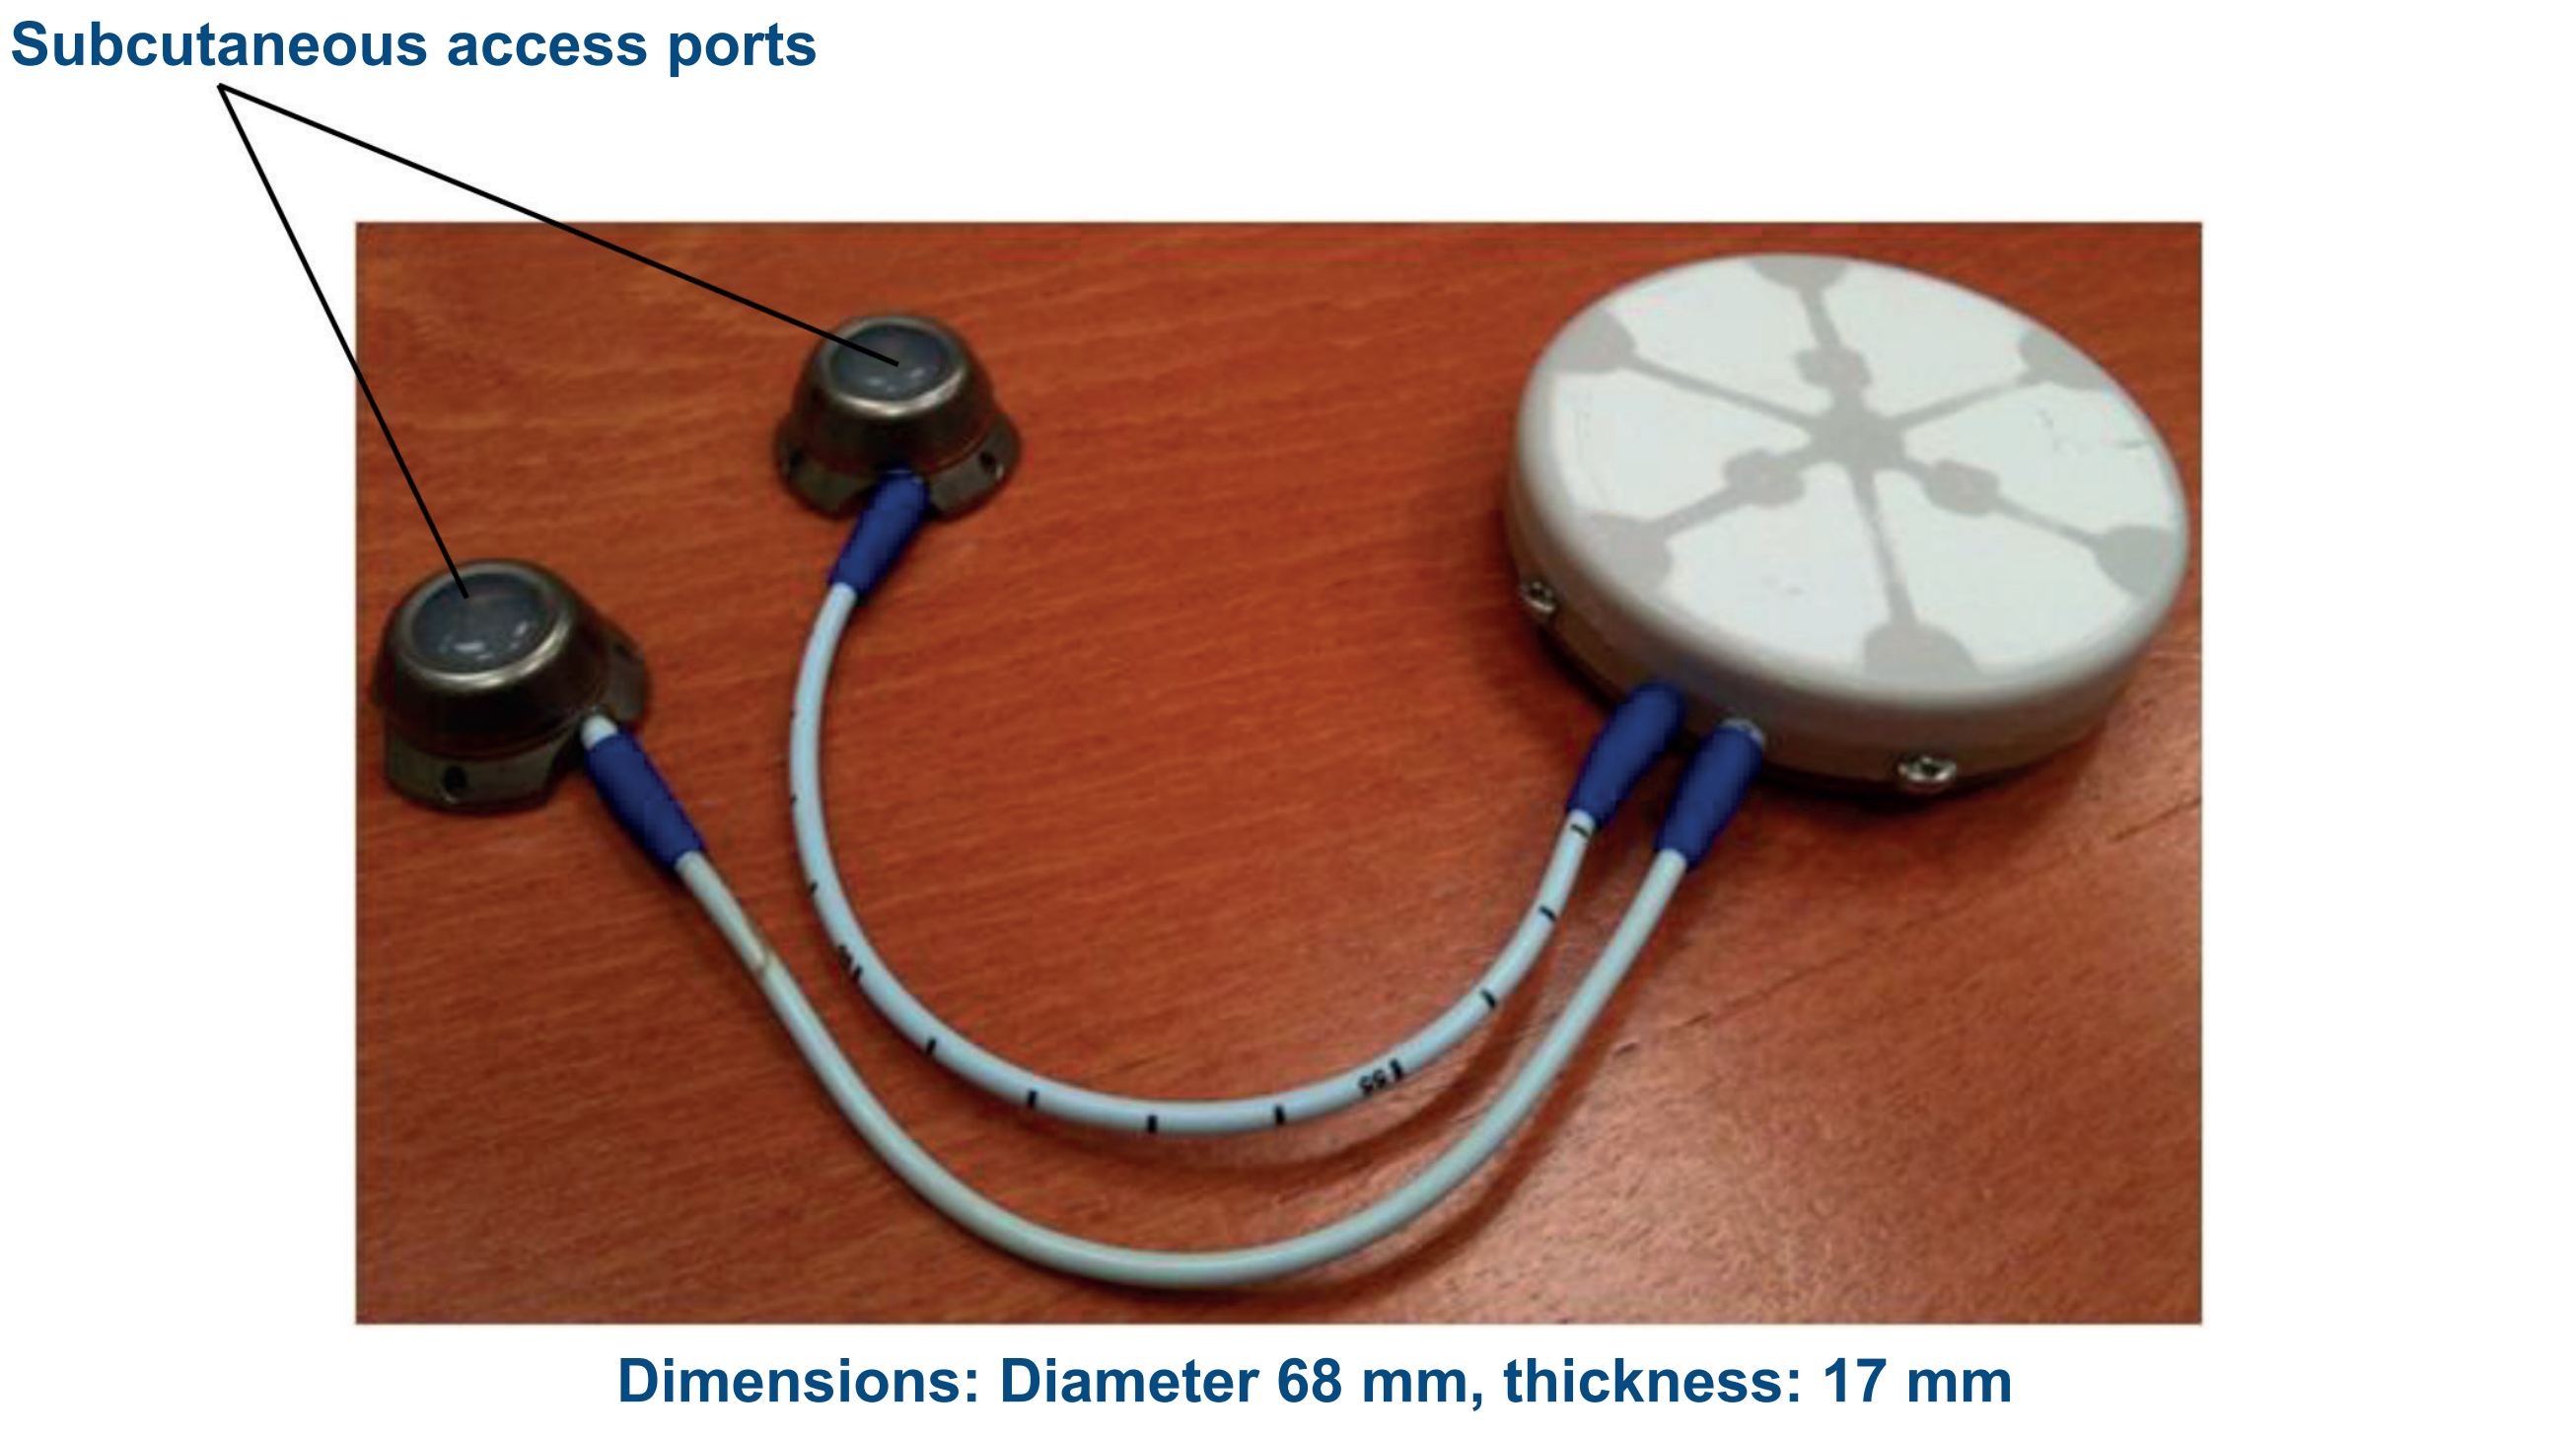

Subcutaneous access ports
Dimensions: Diameter 68 mm, thickness: 17 mm

Figure 2. Pig βAir device showing the macro-chamber containing the islets and the access subcutaneous ports used for O_2 replenishing.

Supplying O_2 to the dense islet culture is key to its continued function and longevity. The O_2 supplied through the access ports crosses the gas permeable membrane, dissolves and diffuses in the gel, then consumed by the islets. The gradient in O_2 partial pressure across the islet culture depends on the density of the culture. Increased density leads to rapid drop in O_2 partial pressure across the culture **(Figure 3)**.

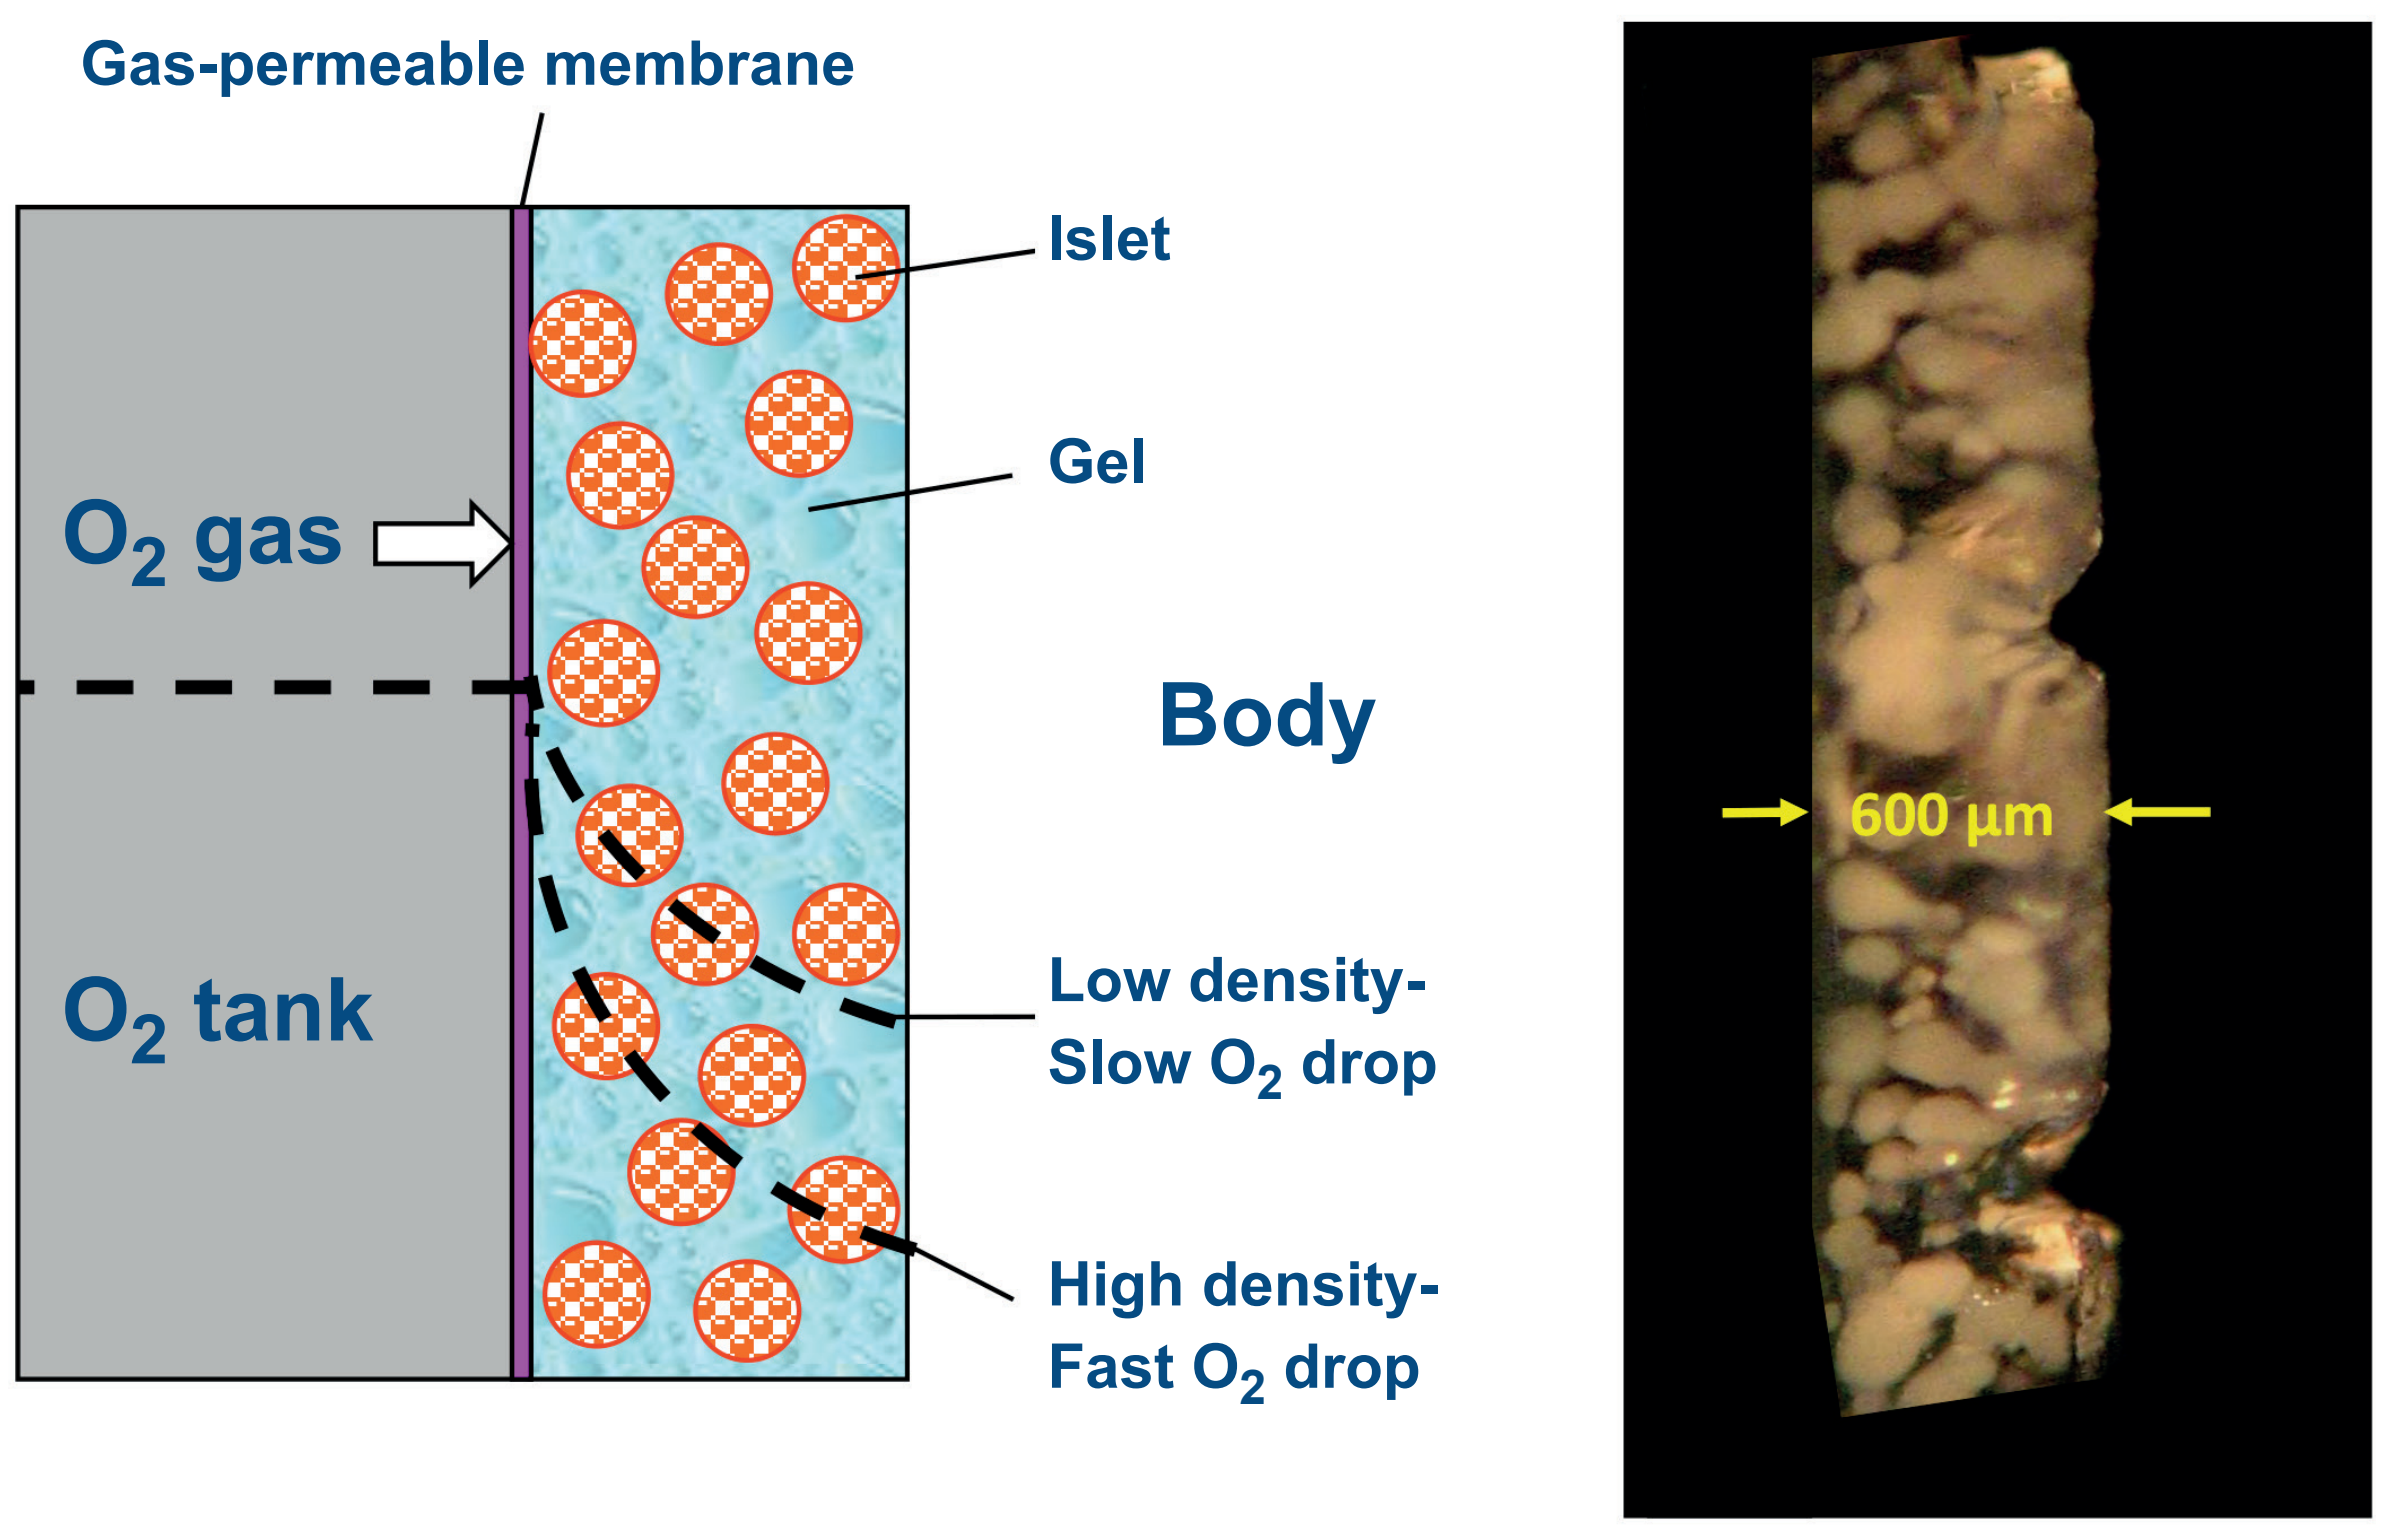

Gas-permeable membrane
Islet
Gel
O2 gas
Body
O2 tank
Low density-
Slow O2 drop
High density-
Fast O2 drop
600 µm

Figure 3. Supplying O_2 to the dense islet culture. Left: Theoretical cross-section of the device, illustrating that increased islet density leads to faster drop in O_2 partial pressure. Right: A cross-section of a highly dense slab (4,800 islet equivalent [IEQ]/cm^2).

Results

The βAir device demonstrated an ability to maintain functionality of the islets with active supply of O_2 in different animal models, and the ability of its membrane to shield donor islets from the host immune system, without immunosuppressive drug therapy.

- βAir devices were implanted in rats with streptozotocin (STZ)-Induced diabetes either with or without replenishing the O_2 supply. Blood glucose levels were measured daily. Without O_2, the βAir device did not affect blood glucose levels **(Figure 4, left)**, whereas with daily O_2 replenishment, near normoglycemia was achieved and sustained until O_2 replenishment was stopped after 59 days **(Figure 4, right)**.

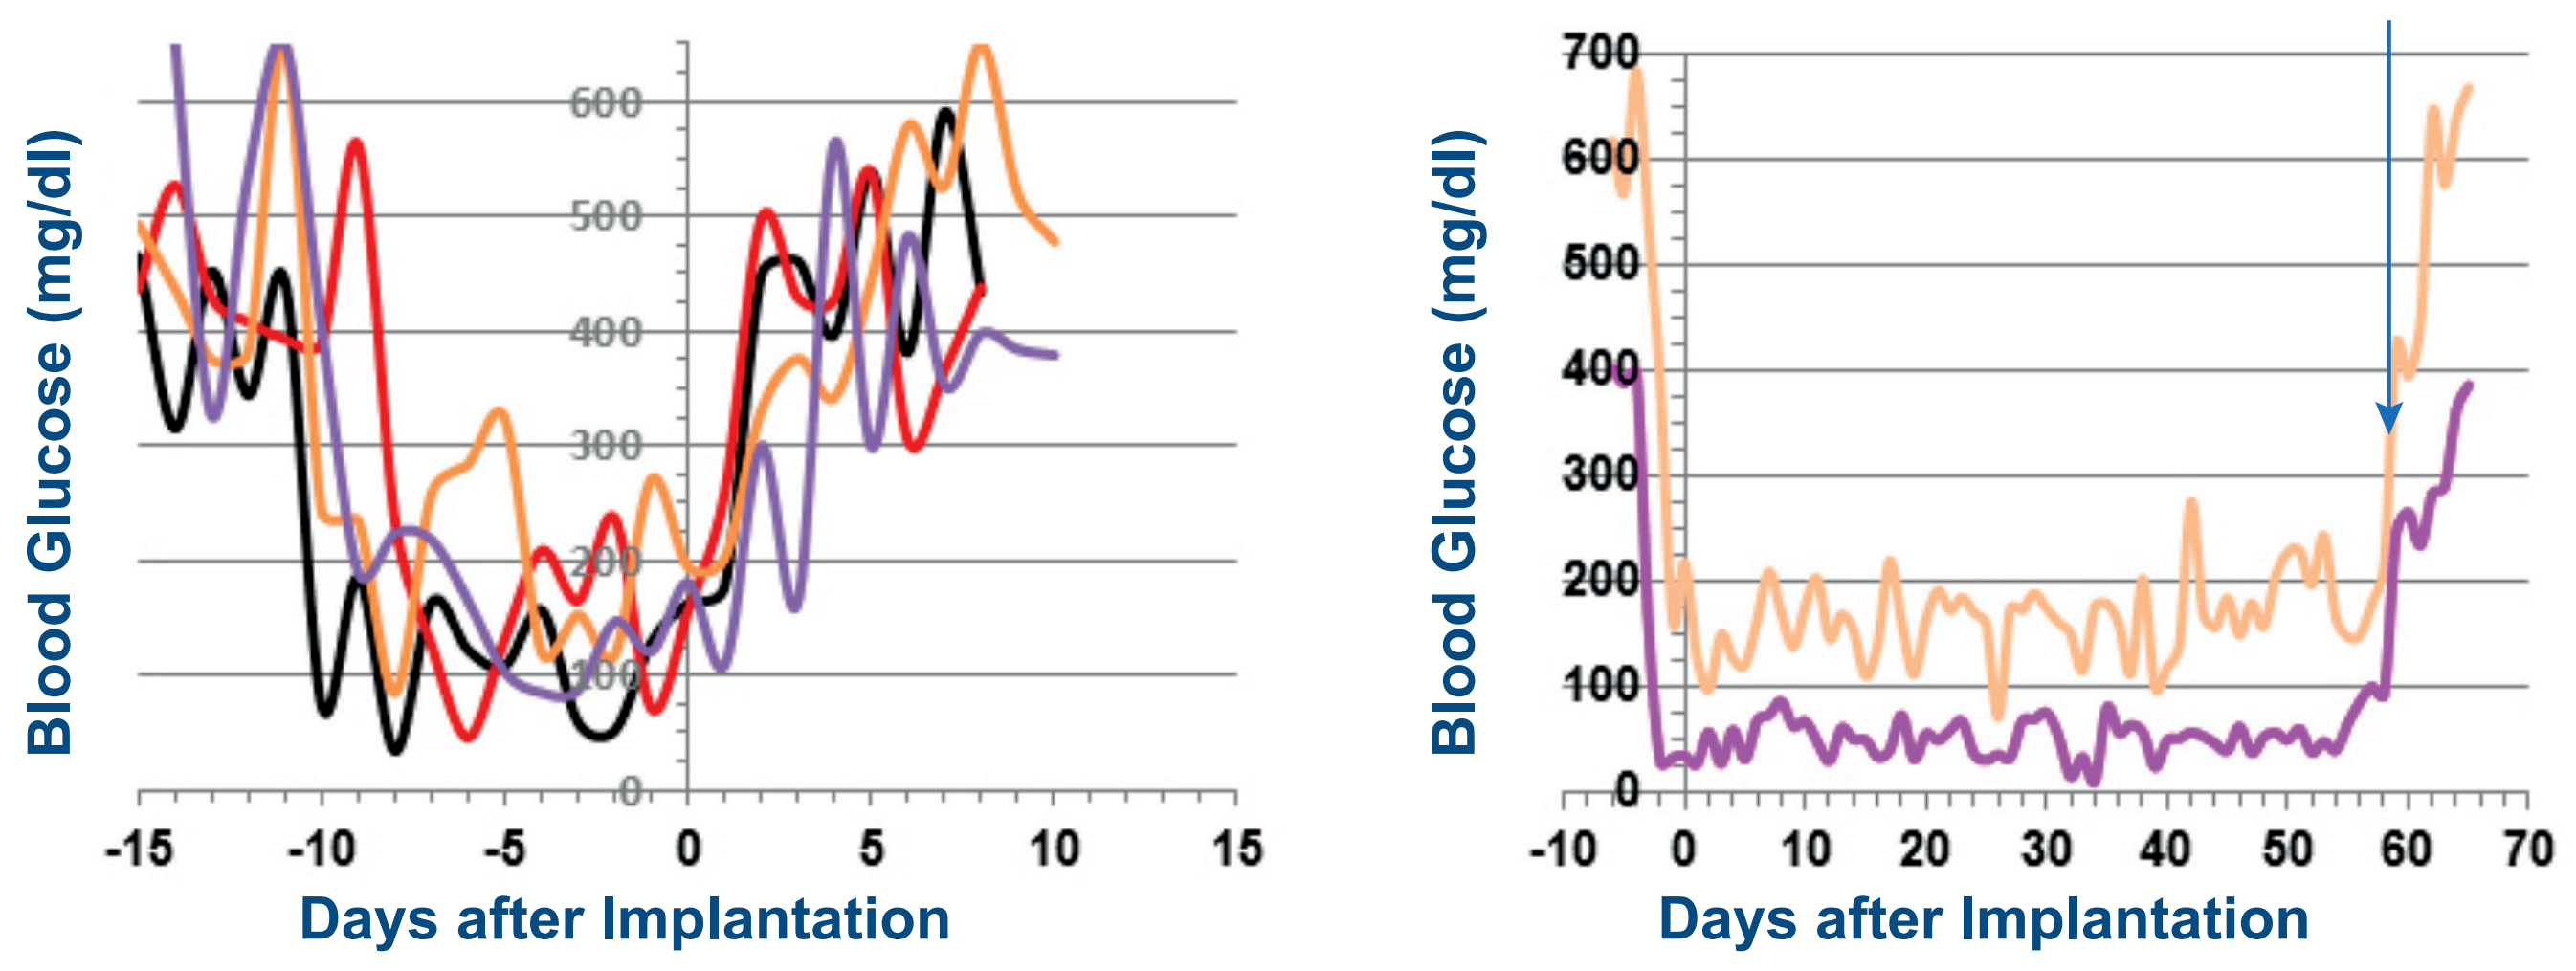

Blood Glucose (mg/dl)
Days after Implantation
Blood Glucose (mg/dl)
Days after Implantation

Figure 4. βAir function is dependent on O_2 supply. Left: STZ-induced diabetic rats (n = 4) were implanted with βAir devices containing rat islets for 9 days with no O_2 replenishment. Right: βAir devices were implanted in STZ-induced diabetic rats (n = 6), O_2 was replenished daily for 58 days, and on day 59, O_2 supply was replaced with N_2. The lines represent 99% upper and lower confidence.

- βAir devices containing rat islets were implanted in isogenic, allogeneic, and xenogeneic animals with STZ-Induced diabetes. Upon implantation, near normoglycemia was achieved. Upon retrieval, blood glucose levels returned to the diseased state **(Figure 5)**.

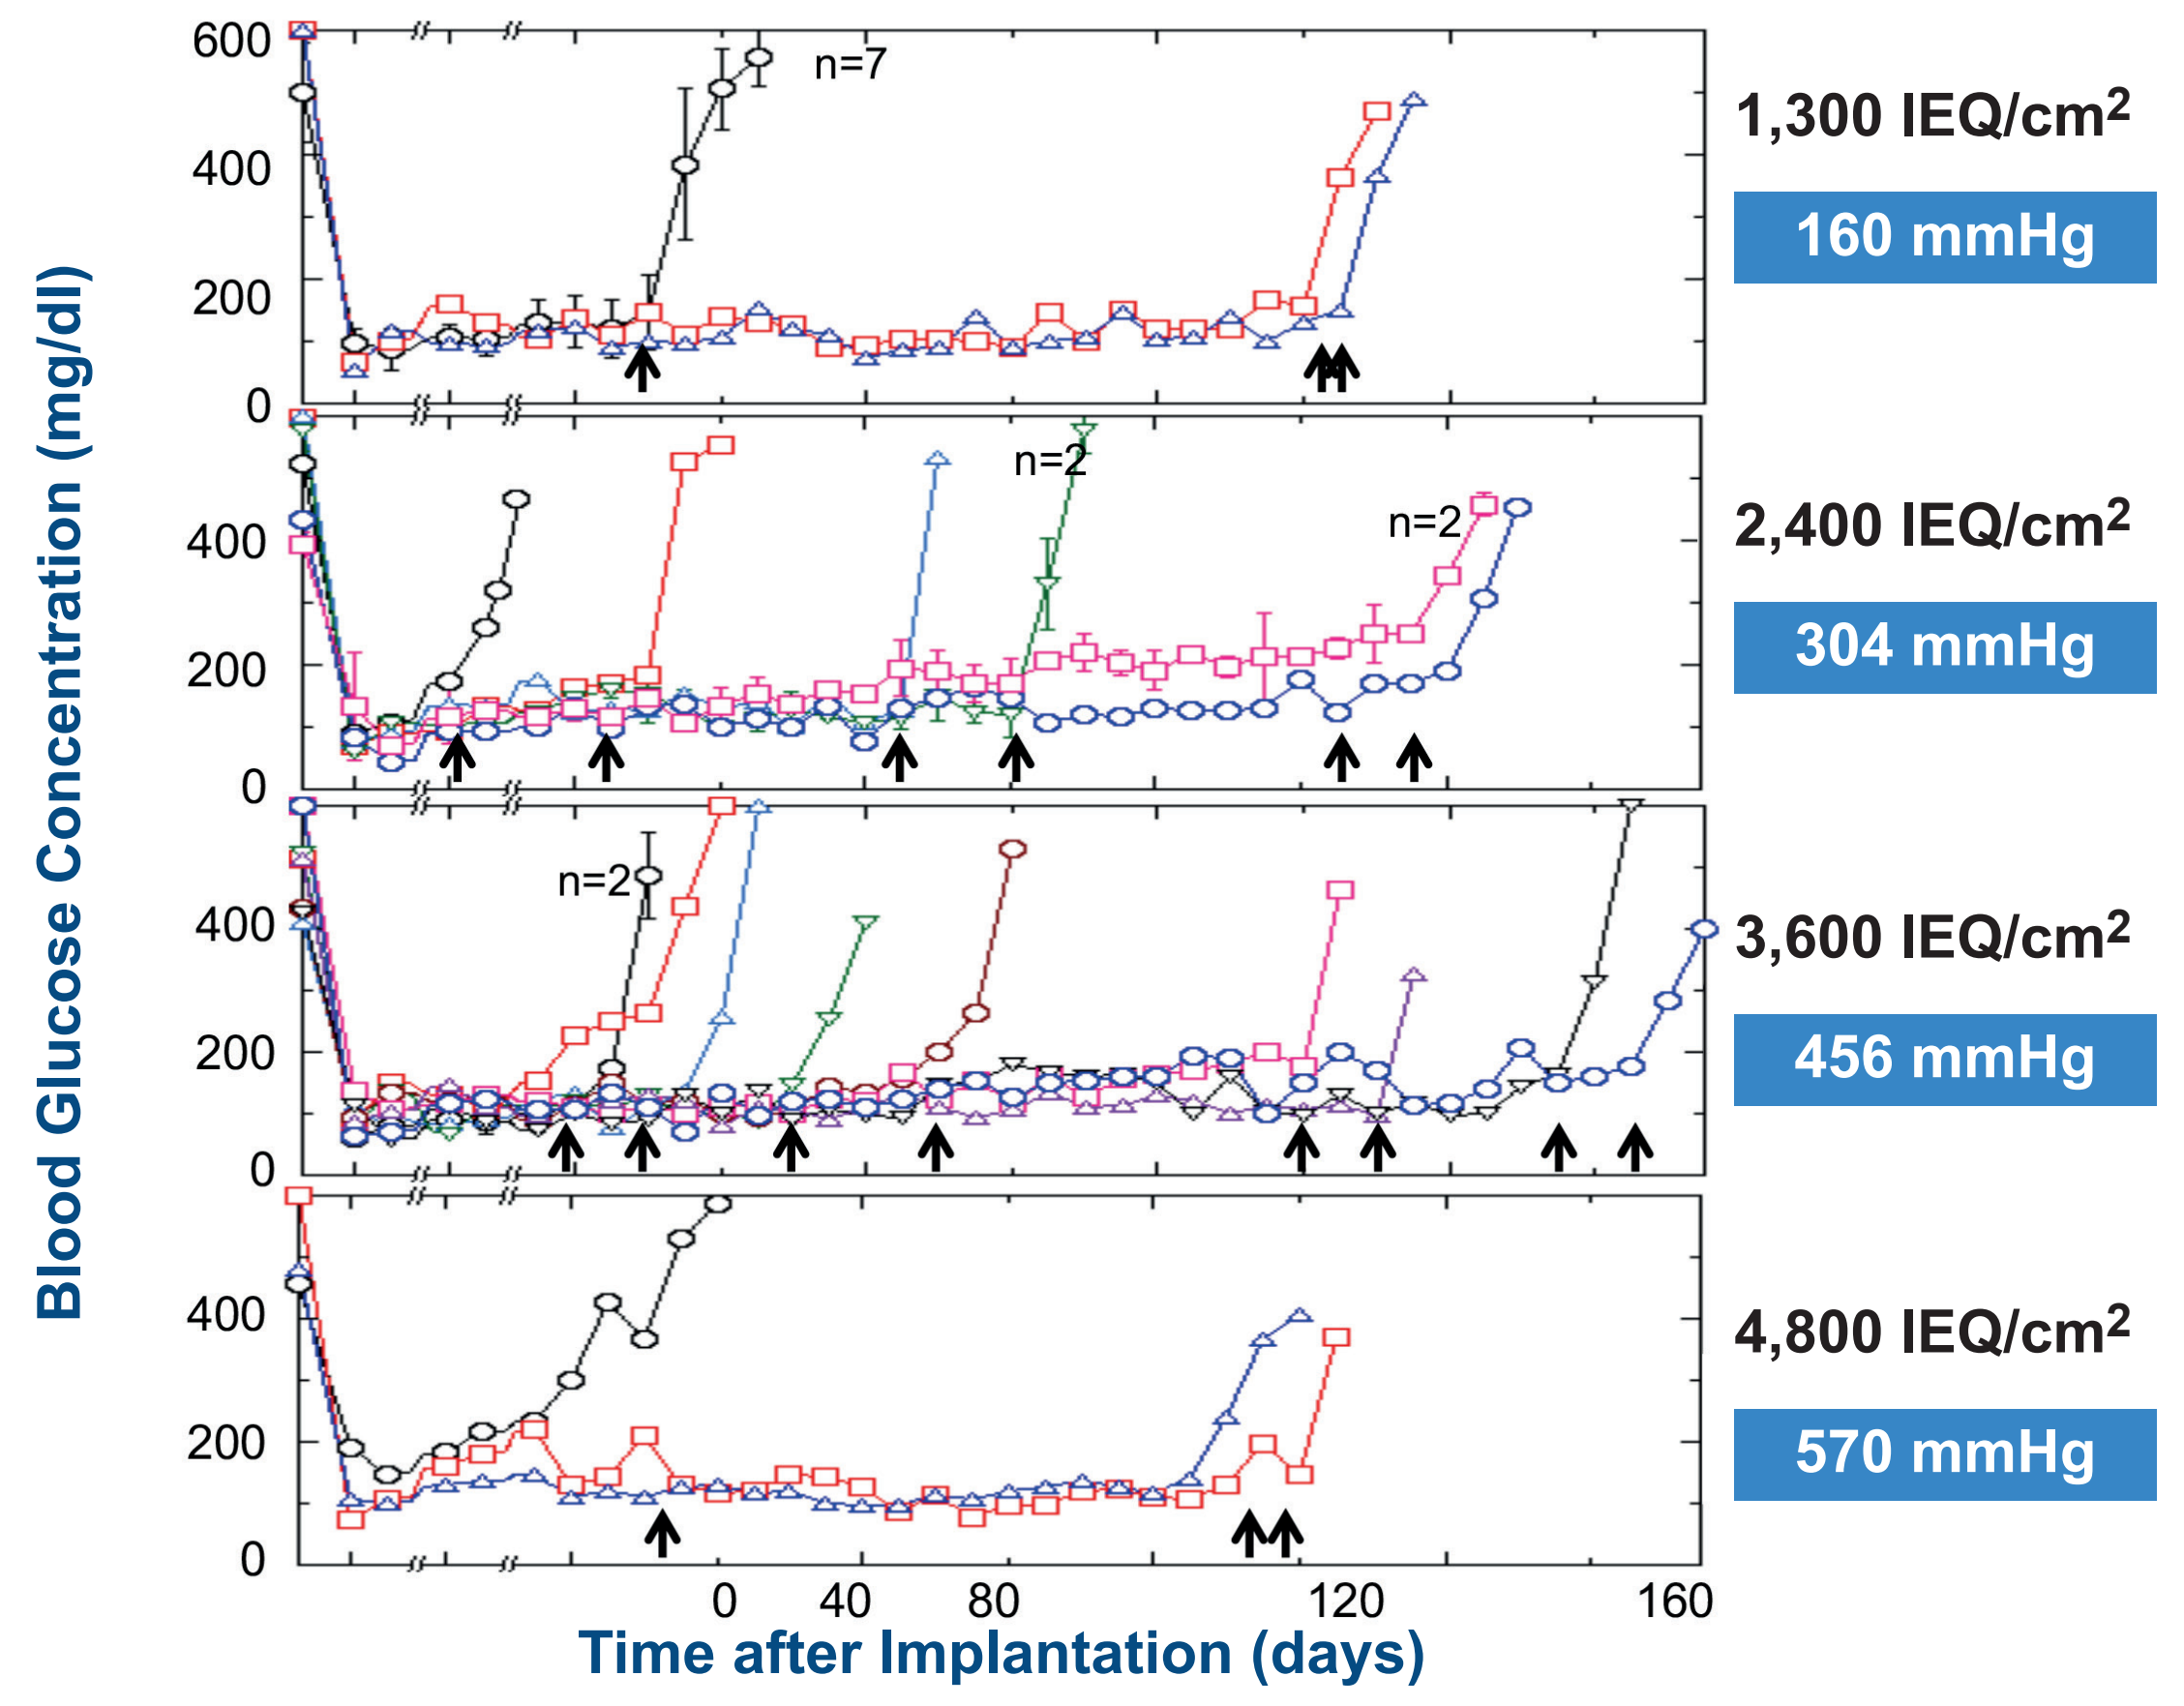

Blood Glucose Concentration (mg/dl)
Time after Implantation (days)
n=7
n=2
n=2
n=2
1,300 IEQ/cm2
160 mmHg
2,400 IEQ/cm2
304 mmHg
3,600 IEQ/cm2
456 mmHg
4,800 IEQ/cm2
570 mmHg

Figure 5. Rat βAir devices containing ~2,400 IEQ/device at different densities and supplied with various O_2 concentrations (blue highlights) were implanted in STZ-induced diabetic rats. Each line represents blood glucose concentrations of another animal, except when otherwise indicated. Arrows represent time of elective retrieval of the device.

- Pig devices containing islets from Lewis rats were implanted in mini-pigs with STZ-induced diabetes. The gas mixture (21-95% O_2, 5% CO_2 and the rest N_2) was daily replenished by trans-cutaneous injection via the access ports, and blood glucose levels were measured. During the implantation period, near normoglycemia was achieved and weight gain was observed. After explantation, blood glucose levels returned to the diseased state and the animals started losing weight **(Figure 6)**.

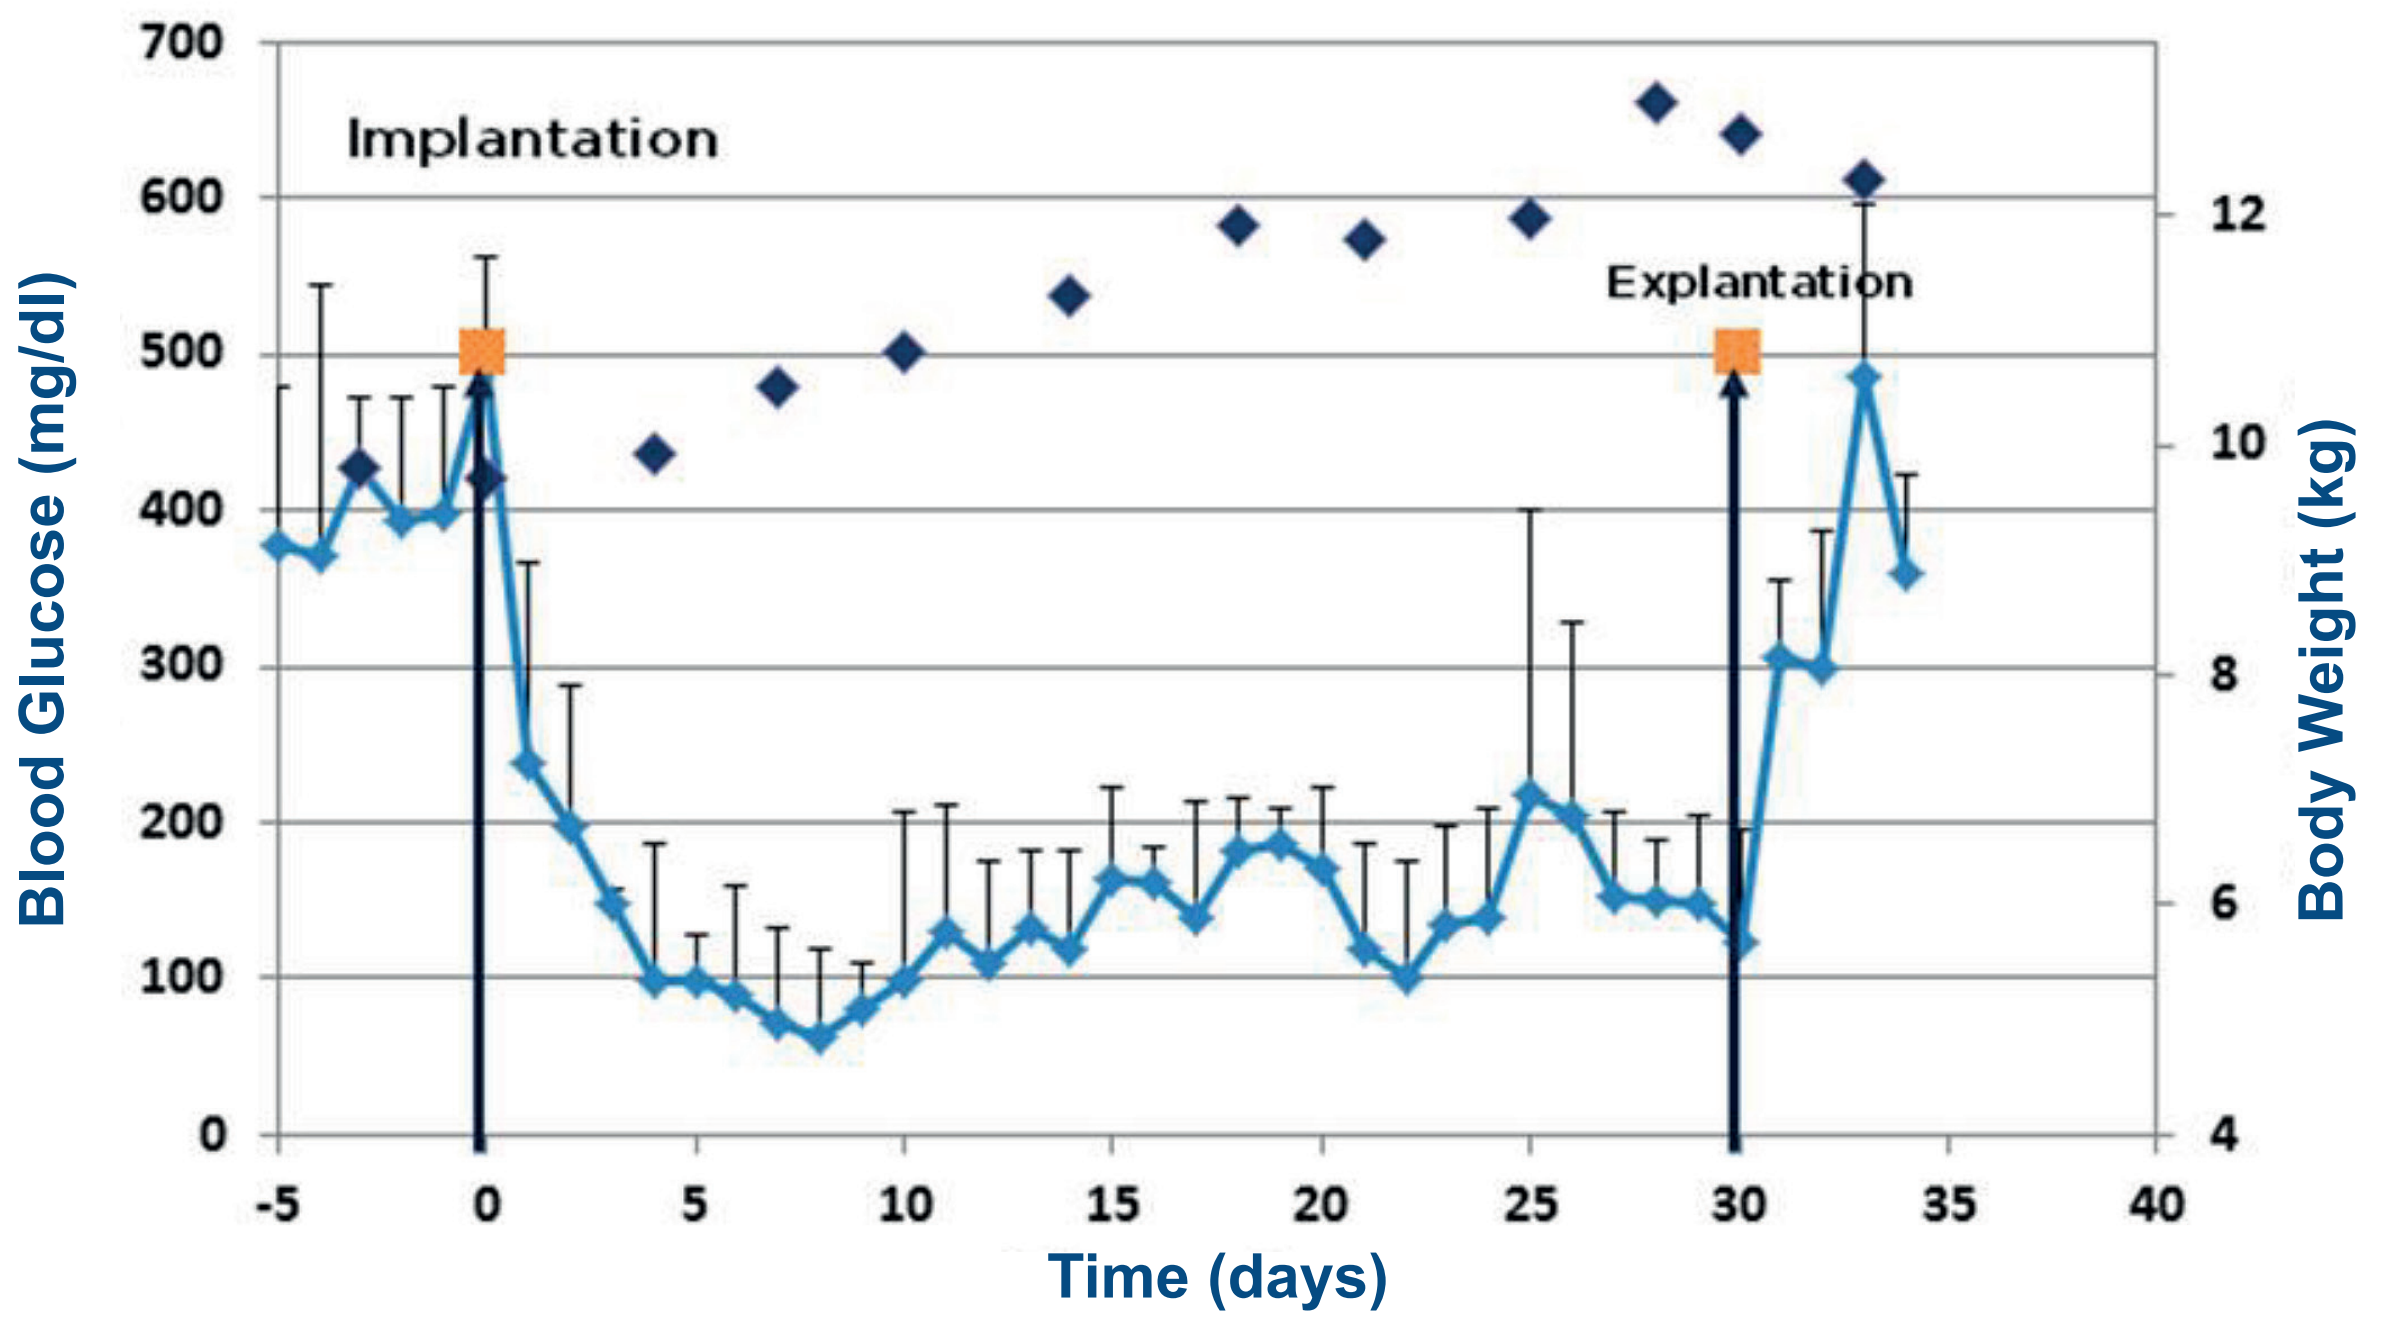

Implantation
Explantation
Blood Glucose (mg/dl)
Body Weight (kg)
Time (days)

Figure 6. Pig βAir devices containing ~6,500 IEQ/Kg body weight were implanted in STZ-induced diabetic mini-pigs (n = 4). Light blue represents mean blood glucose levels with SD. Blue diamonds represent the mean weight of the animals.

Conclusions

Preclinical results in 3 animal models proved the ability of the βAir device to:

- **Support efficient replenishing with O_2 for continued function of the donor islets at different densities**
- **Protect isogenic, allogenic, and xenogeneic implanted cells from the host immune system without any immunosuppressive drugs**
- **Achieve near-normal glucose levels in 3 diabetic animals models**

References

1. Barkai U et al. Enhanced oxygen supply improves islet viability in a new bioartificial pancreas. Cell Transpl. 22: 1463-1476 (2013).
2. Neufeld T et al. The efficacy of an immunoisolating membrane system for islet xenotransplantation in minipigs. PLoS One 8: e70150 (2013).